A host defense peptide mimetic, brilacidin, potentiates caspofungin antifungal activity against human pathogenic fungi.

Thaila Reis[1], Patrícia A. de Castro[1], Rafael W. Bastos[1], Camila F. Pinzan[1], Pedro F. N. Souza[2], Suzanne Ackloo[3], Mohammad A. Hossain[4], David H. Drewry[4], Sondus Alkhazraji[5], Ashraf S. Ibrahim[5], Hyunil Jo[6], Jorge D. Lightfoot[7], Emily M. Adams[7], Kevin K. Fuller[7], William F. DeGrado[6], Gustavo H. Goldman[1]

1) University of Sao Paulo, Ribeirao Preto, Brazil; 2) Federal University of Ceará, Fortaleza, Brazil; 3) University of Toronto, Toronto, Canada; 4) University of North Carolina at Chapel Hill, Chapel Hill, USA; 5) University of California Los Angeles (UCLA), Torrance, USA; 6) University of California, San Francisco, USA; 7) University of Oklahoma Health Sciences Center, Oklahoma, USA.

E-mail: ggoldman@usp.br

ABSTRACT

Fungal infections cause more than 1.5 million deaths a year. Due to emerging antifungal drug resistance, novel strategies are urgently needed to combat life threatening fungal diseases. Here, we identified the host defense peptide mimetic, brilacidin (BRI) as a synergizer with caspofungin (CAS) against CAS sensitive and CAS-resistant isolates of *Aspergillus fumigatus, Candida albicans*, *C. auris*, and CAS-intrinsically resistant *Cryptococcus neoformans*. BRI also potentiates azoles against *A. fumigatus* and several Mucorales fungi. BRI acts in *A. fumigatus* by affecting cell wall integrity pathway and cell membrane potential. BRI combined with CAS significantly clears *A. fumigatus* lung infection in an immunosuppressed murine model of invasive pulmonary aspergillosis. BRI alone also decreases *A. fumigatus* fungal burden and ablates disease development in a murine model of fungal keratitis. Our results indicate that combinations of BRI and antifungal drugs in clinical use are likely to improve the treatment outcome of aspergillosis and other fungal infections.

RESULTS

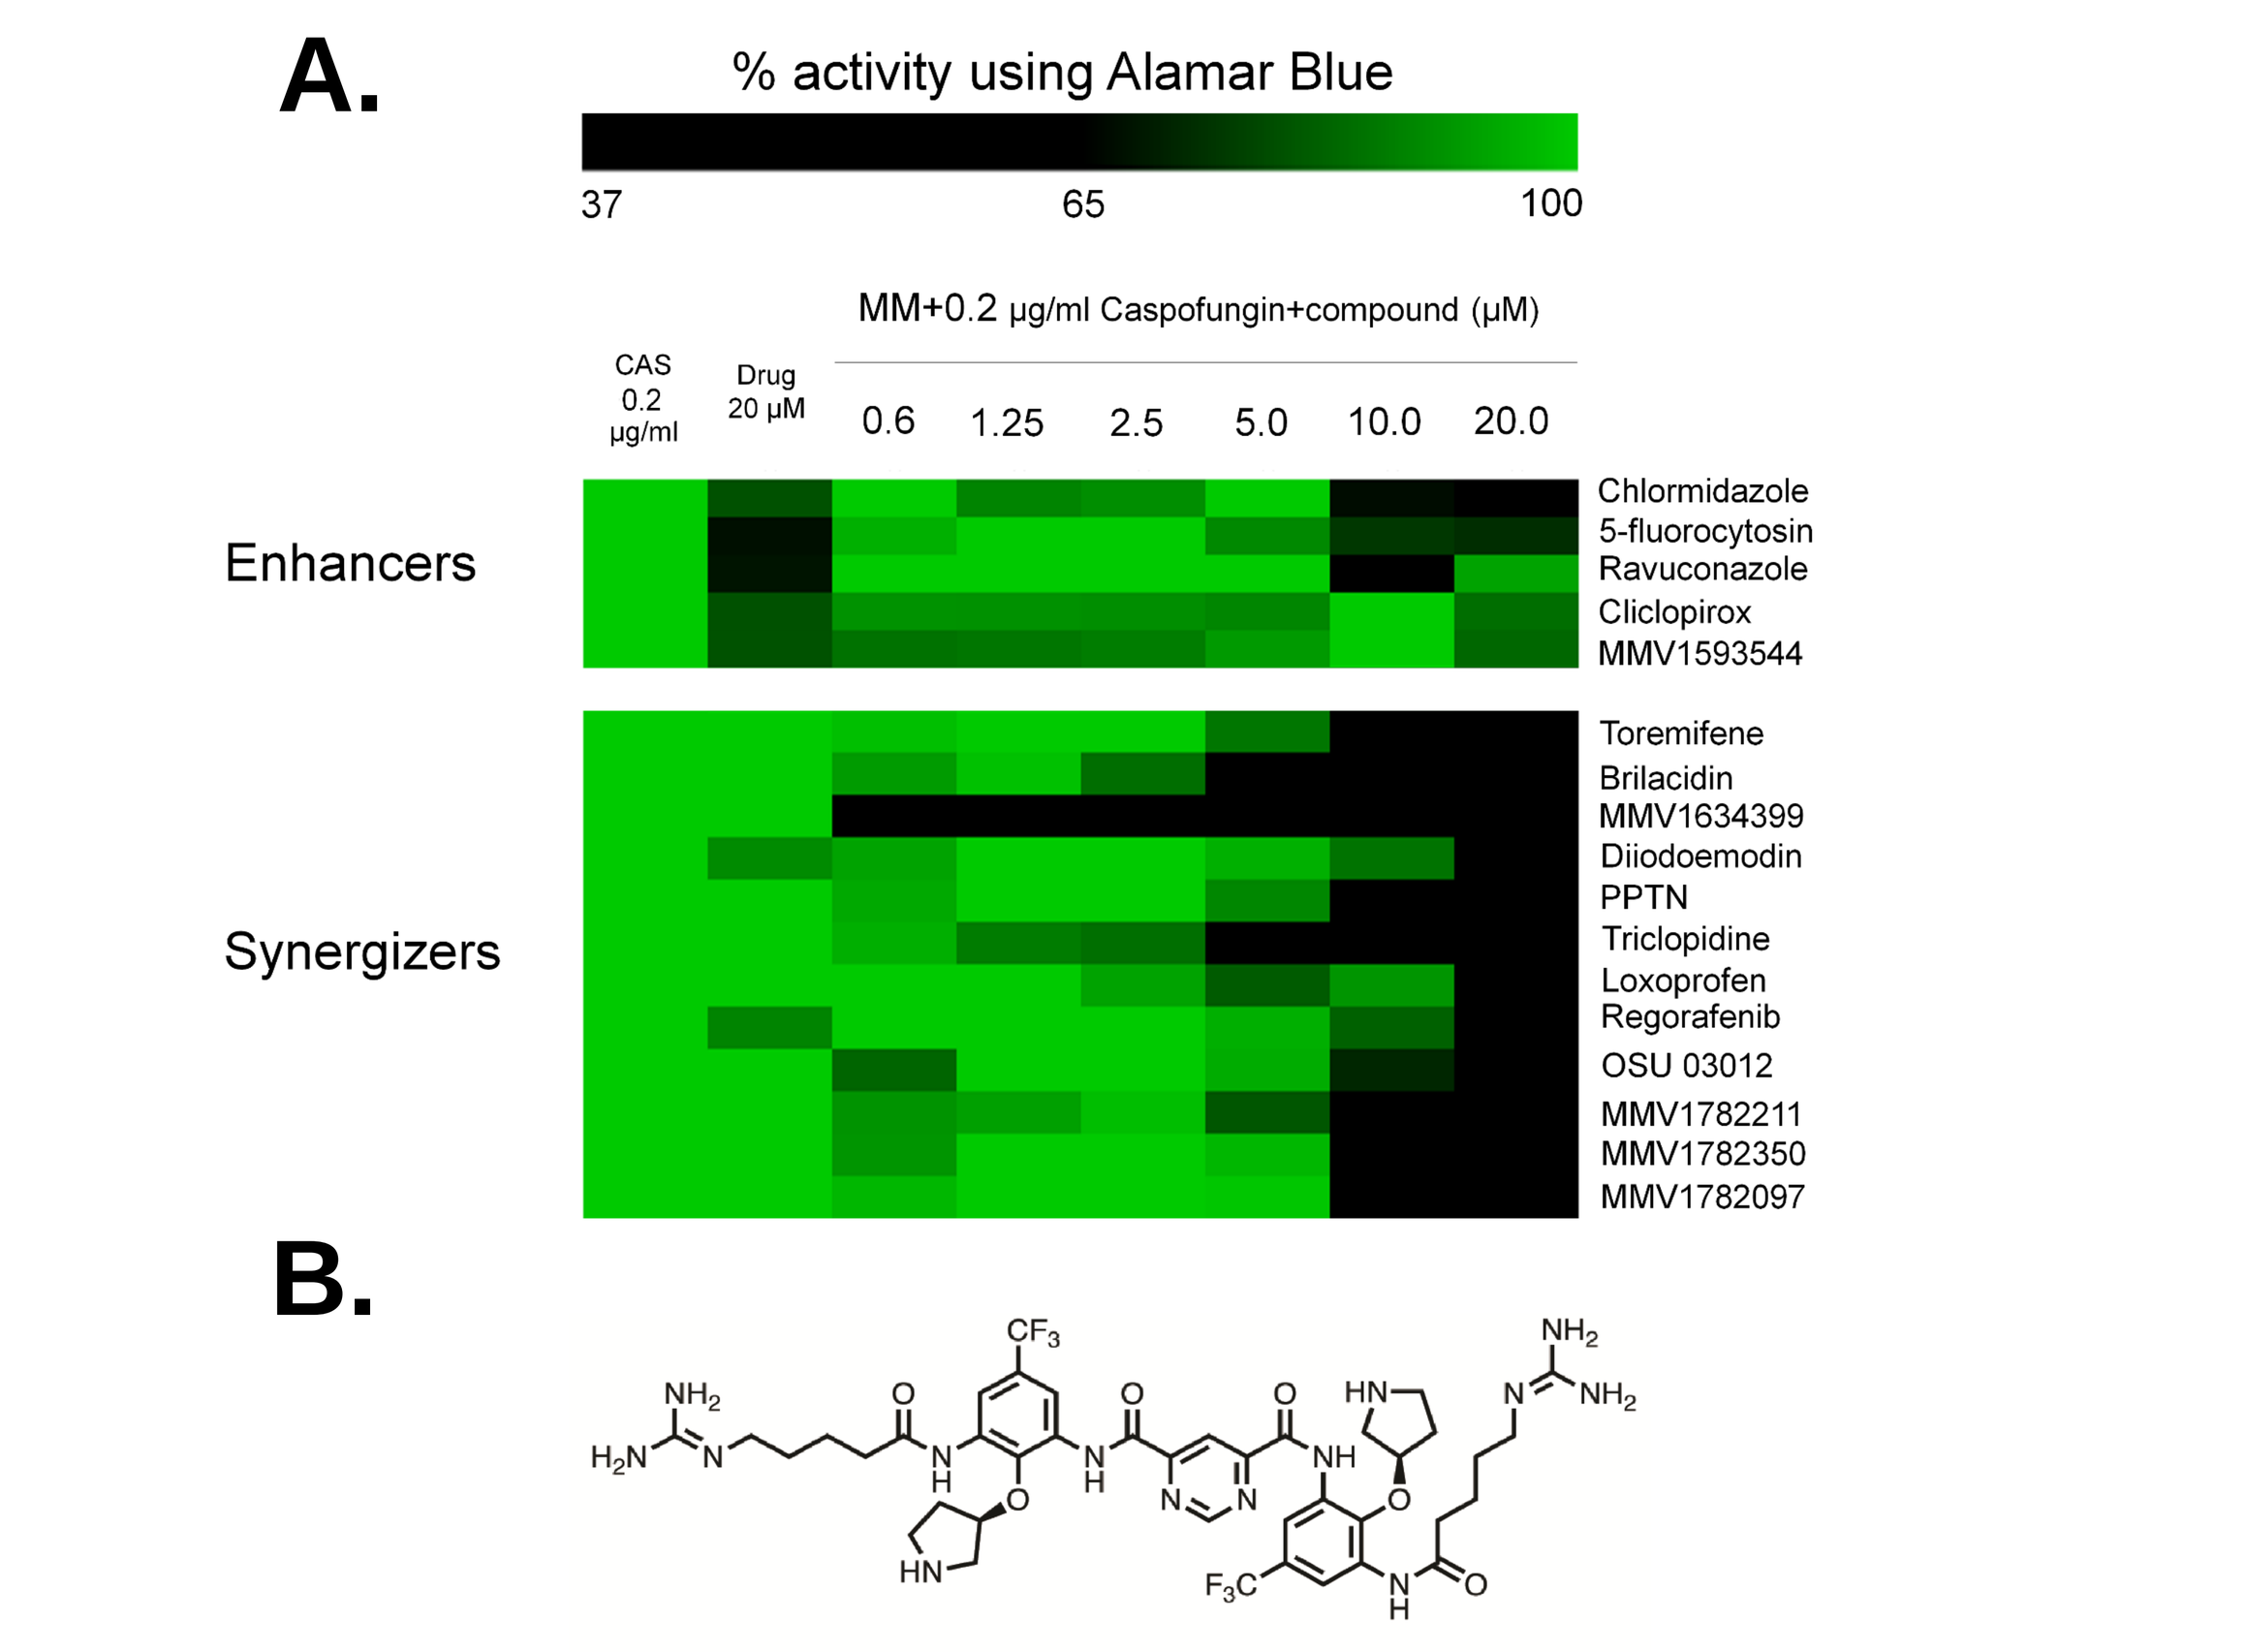

A.
% activity using Alamar Blue
37
65
100
MM+0.2 µg/ml Caspofungin+compound (µM)
CAS 0.2 µg/ml
Drug 20 µM
0.6 1.25 2.5 5.0 10.0 20.0
Enhancers
Chlormidazole
5-fluorocytosin
Ravuconazole
Ciclopirox
MMV1593544
Synergizers
Toremifene
Brilacidin
MMV1634399
Diiodoemodin
PPTN
Triclopidine
Loxoprofen
Regorafenib
OSU 03012
MMV1782211
MMV1782350
MMV1782097
B.

Brilacidin (MMV1634402)

Figure 1. Screening of chemical libraries identify compounds that enhance or synergize caspofungin activity. (A) Heat map of % of metabolic activity using Alamar blue. The % of activity is based on *A. fumigatus* grown for 48h at 37°C in (i) minimal medium (MM) or (ii) MM containing CAS 0.2 µg/mL, 20µM of chemical compound alone, or a combination of CAS and chemical compound(from 0.6 to 20 µM) divided by the control (MM), both grown for 48h at 37°C. **(B)** Chemical structure of BRI.

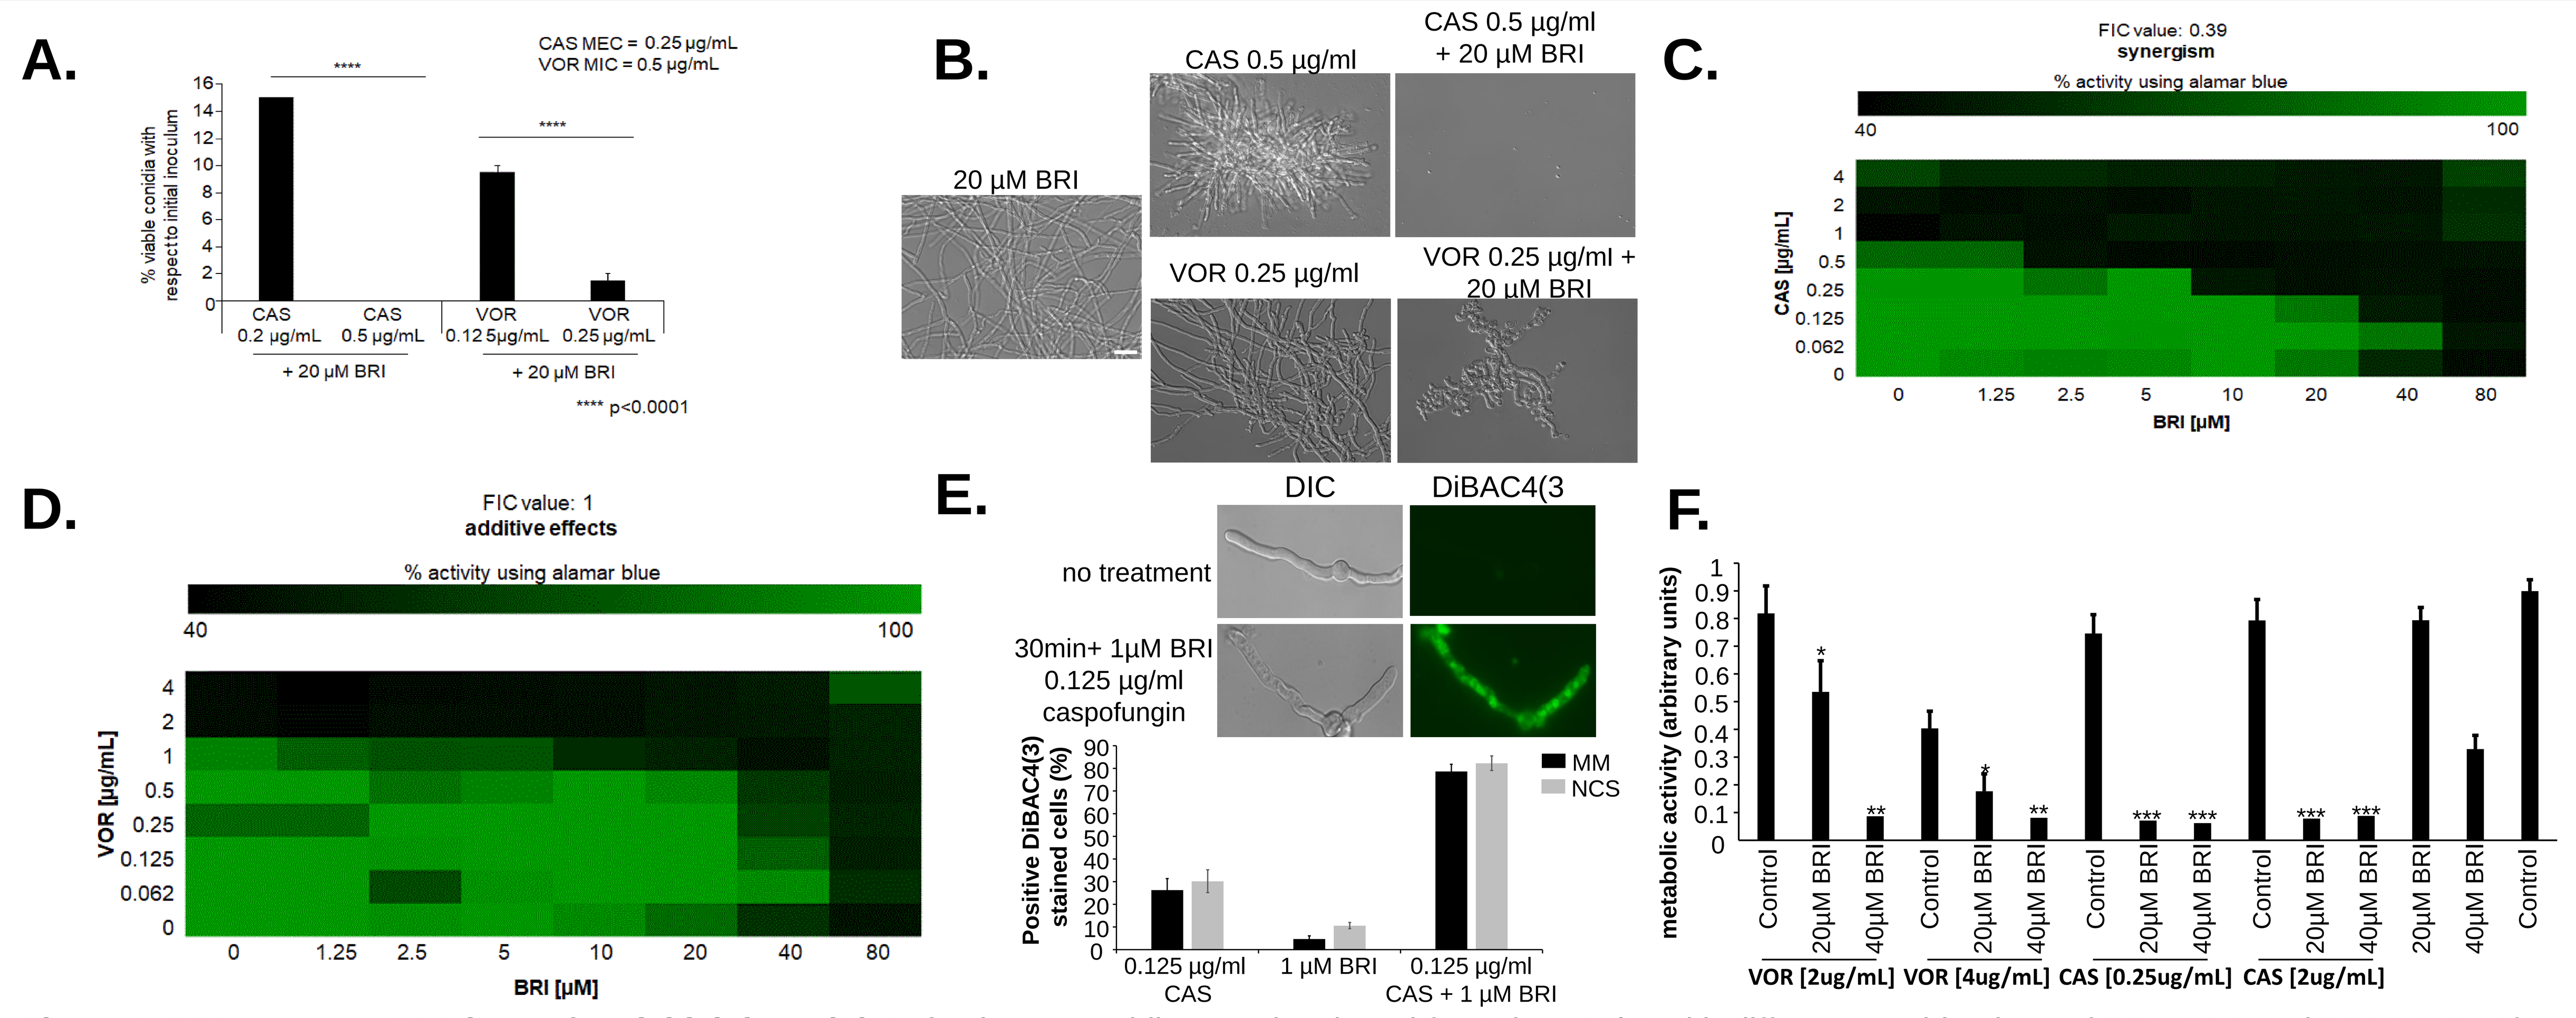

A.
CAS MEC = 0.25 µg/mL
VOR MIC = 0.5 µg/mL
% viable conidia with respect to initial inoculum
CAS 0.2 µg/mL
CAS 0.5 µg/mL
VOR 0.12 5µg/mL
VOR 0.25 µg/mL
+ 20 µM BRI
**** p<0.0001
B.
20 µM BRI
CAS 0.5 µg/ml
CAS 0.5 µg/ml + 20 µM BRI
VOR 0.25 µg/ml
VOR 0.25 µg/ml + 20 µM BRI
C.
FIC value: 0.39
synergism
% activity using alamar blue
CAS [µg/mL]
BRI [µM]
D.
FIC value: 1
additive effects
% activity using alamar blue
VOR [µg/mL]
BRI [µM]
E.
DIC
DiBAC4(3
no treatment
30min+ 1µM BRI 0.125 µg/ml caspofungin
Positive DiBAC4(3) stained cells (%)
MM
NCS
0.125 µg/ml CAS
1 µM BRI
0.125 µg/ml CAS + 1 µM BRI
F.
metabolic activity (arbitrary units)
VOR [2ug/mL]
VOR [4ug/mL]
CAS [0.25ug/mL]
CAS [2ug/mL]

Figure 2. BRI converts CAS into a fungicidal drug. (A) *A. fumigatus* conidia were incubated for 48h at 37°C with different combinations of BRI+CAS and BRI+VOR. After, non-germinated conidia were plated on MM and colony forming units (CFUs) were assessed. The results are expressed as the % of viable conidia with respect to initial inoculum and are the average of three repetitions ± standard deviation. **(B)** Microscopic images of *A. fumigatus* after 48h exposure to BRI, CAS, VOR, BRI + CAS, and BRI + VOR. Bars, 20 µm. **(C)** and **(D)** The Fractional Inhibitory Concentration (FIC) index for BRI+CAS and BRI+VOR, respectively. **(E)** BRI+CAS disrupts the *A. fumigatus* membrane potential. *A. fumigatus* was grown for 16h at 37°C in MM and exposed 4h to MM or MM without glucose (non-carbon source, NCS) containing CAS or CAS + BRI for 30 min and 3µg/mL DIBAC4(3). The results show the % of fluorescent cells and are the average of three repetitions of 50 germlings each ± standard deviation. **(F)** Metabolic activity expressed by XTT of *A. fumigatus* 24h old formed biofilm at 37°C and further treated with VOR, CAS, BRI+VOR or BRI+CAS for 12h at 37°C. Untreated biofilm was used as a positive control. The XTT assays were performed in six replicates. The results are the average of six repetitions ± standard deviation.* (p < .05), ** (p < .01) *** (p < .001) **** (p <.0001).

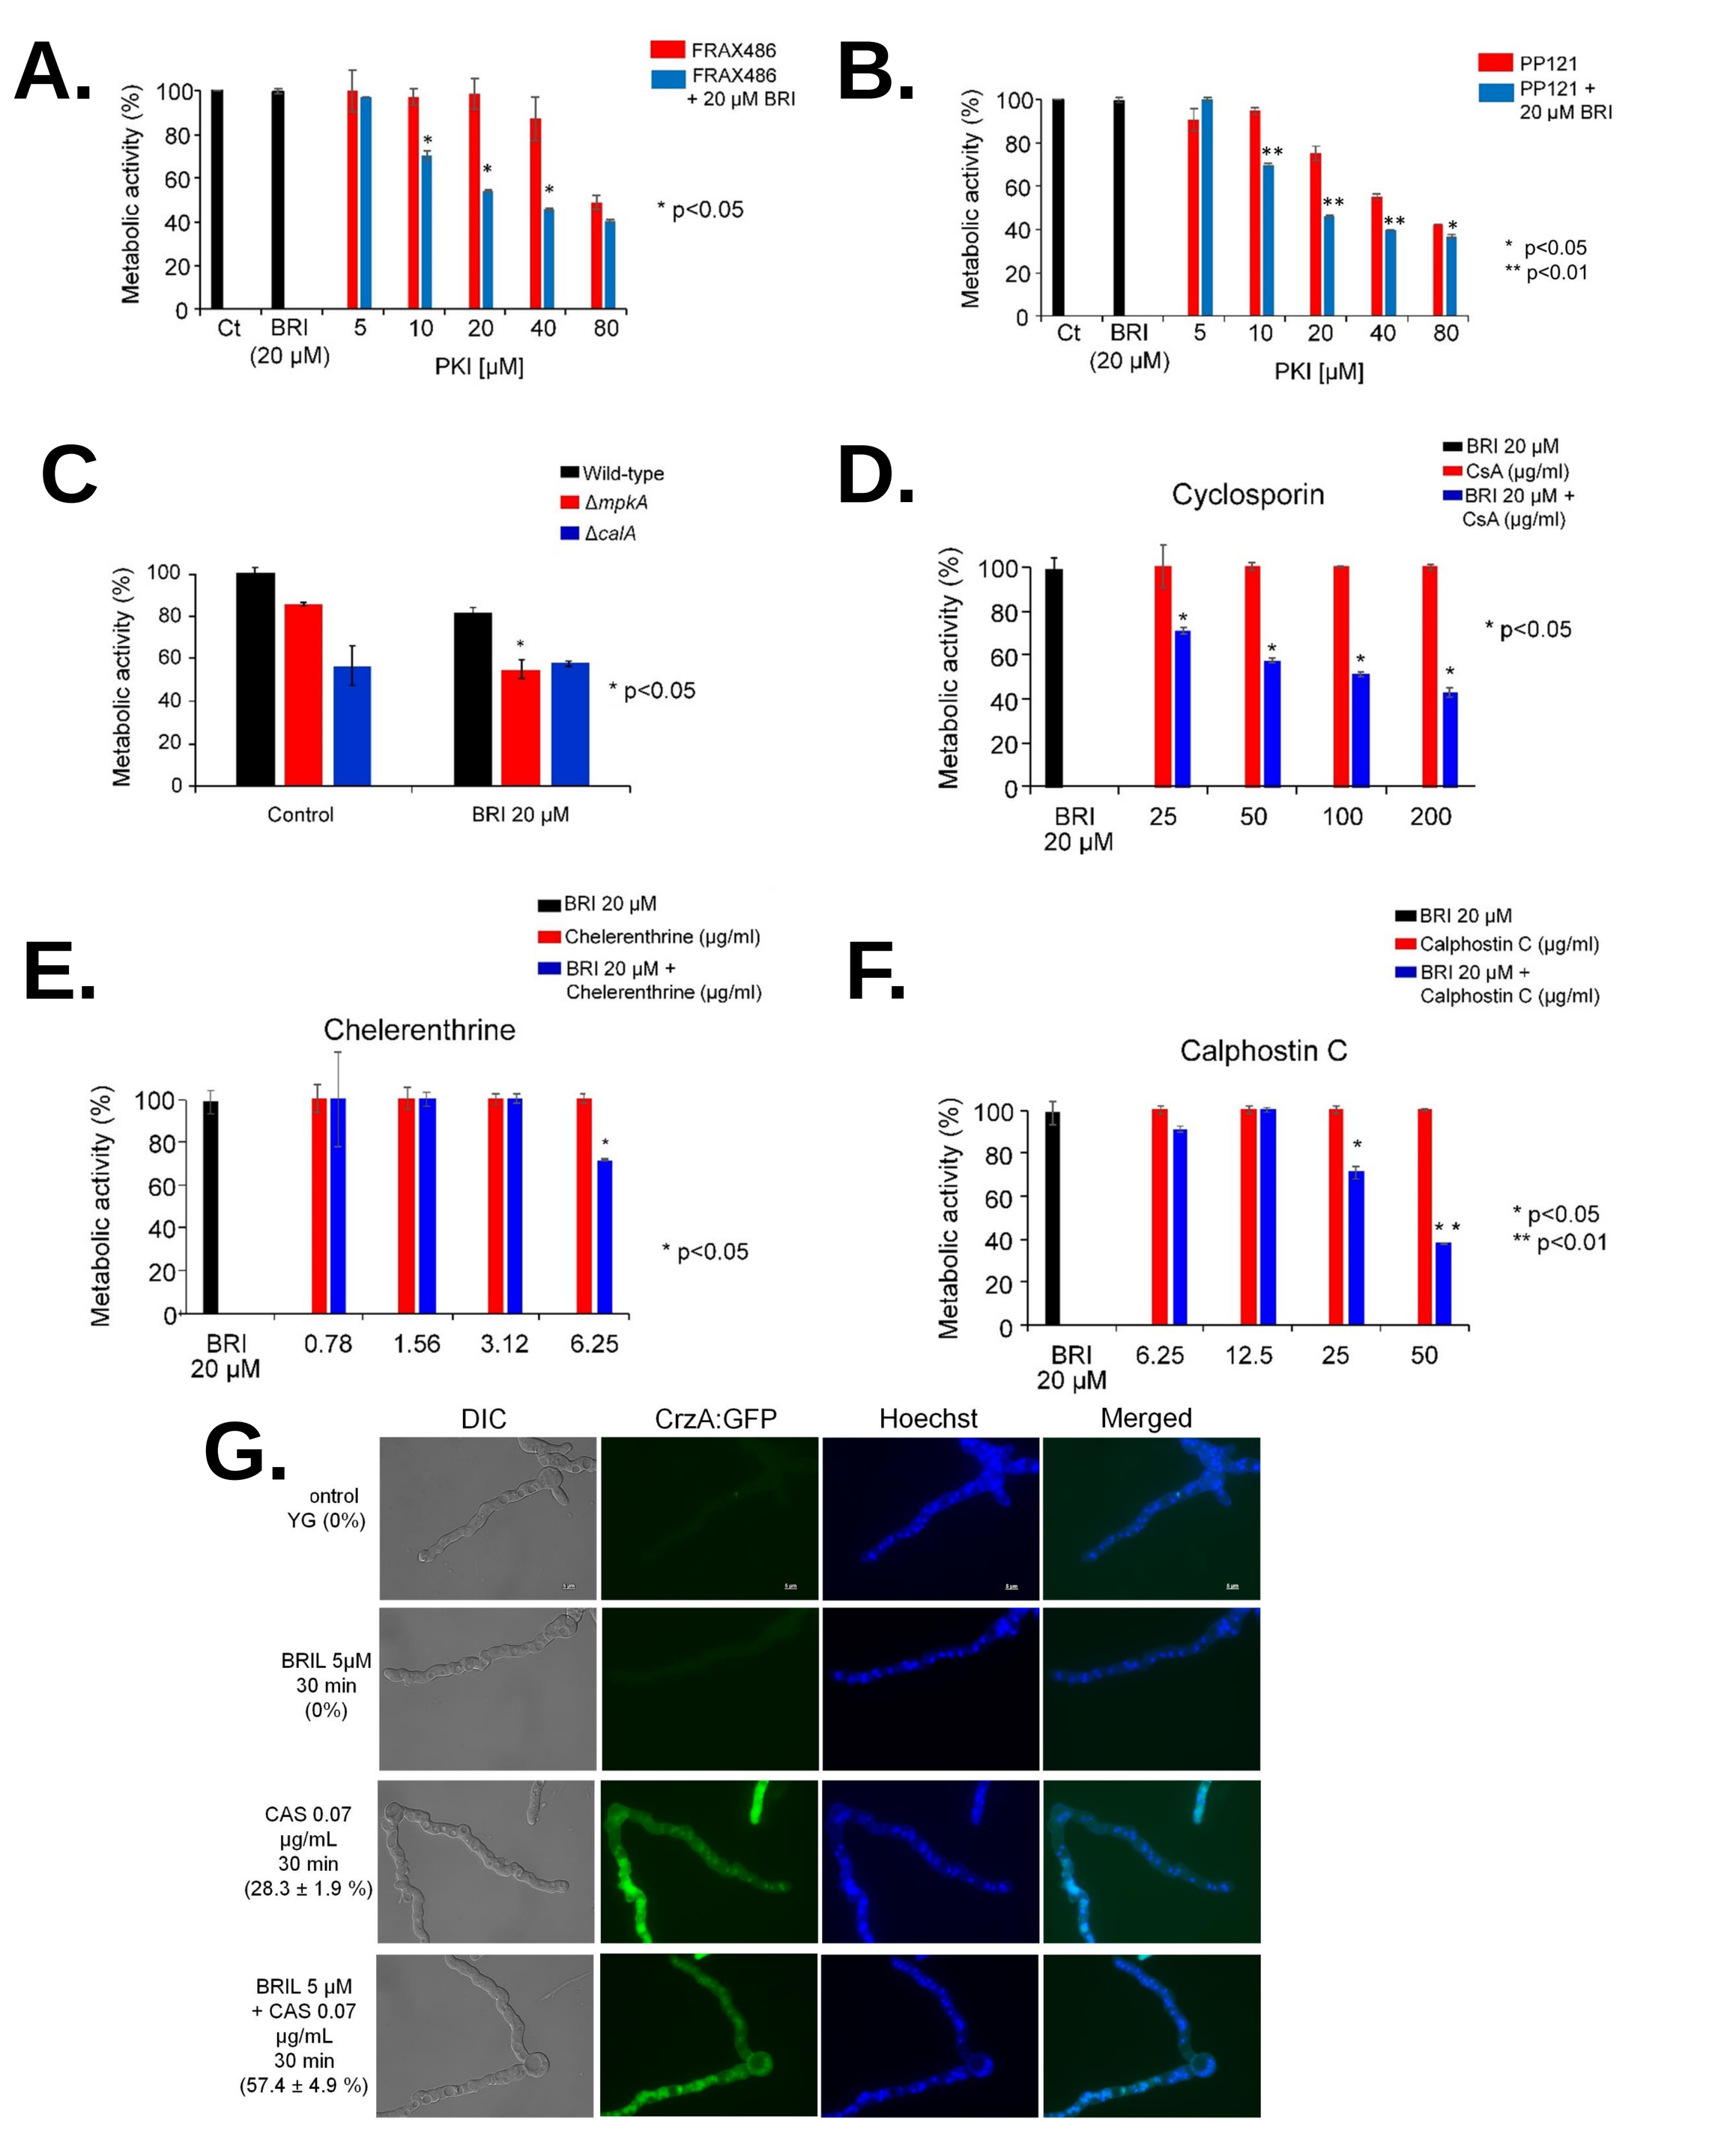

A.
FRAX486
FRAX486 + 20 µM BRI
Metabolic activity (%)
PKI [µM]
* p<0.05
B.
PP121
PP121 + 20 µM BRI
* p<0.05
** p<0.01
C
Wild-type
ΔmpkA
ΔcalA
Control
BRI 20 µM
* p<0.05
D.
Cyclosporin
BRI 20 µM
CsA (µg/ml)
BRI 20 µM + CsA (µg/ml)
* p<0.05
E.
Chelerenthrine
BRI 20 µM
Chelerenthrine (µg/ml)
BRI 20 µM + Chelerenthrine (µg/ml)
* p<0.05
F.
Calphostin C
BRI 20 µM
Calphostin C (µg/ml)
BRI 20 µM + Calphostin C (µg/ml)
* p<0.05
** p<0.01
G.
DIC
CrzA:GFP
Hoechst
Merged
BRIL 5µM 30 min (0%)
CAS 0.07 µg/mL 30 min (28.3 ± 1.9 %)
BRIL 5 µM + CAS 0.07 µg/mL 30 min (57.4 ± 4.9 %)

Figure 3. Calcineurin and the MpkA are important for the BRI+CAS synergism. Metabolic activity expressed by Alamar blue of *A. fumigatus* grown for 48h in the absence or presence of BRI or **(A)** BRI + FRAX486; **(B)** BRI + PP121. **(C)** Metabolic activity expressed by Alamar blue of *A. fumigatus* wild-type, Δ*calA*, and Δ*mpkA* grown for 48h in the absence or presence of BRI. Metabolic activity expressed by Alamar blue of *A. fumigatus* grown for 48h in the absence or presence of BRI or **(D)** BRI + cyclosporin; **(E)** BRI + chelerenthrine; **(F)** BRI + calphostin C. The results are the average of three repetitions ± standard deviation. **(G)** CrzA:GFP translocates to the nucleus when germlings were exposed to BRI, CAS or BRI + CAS.

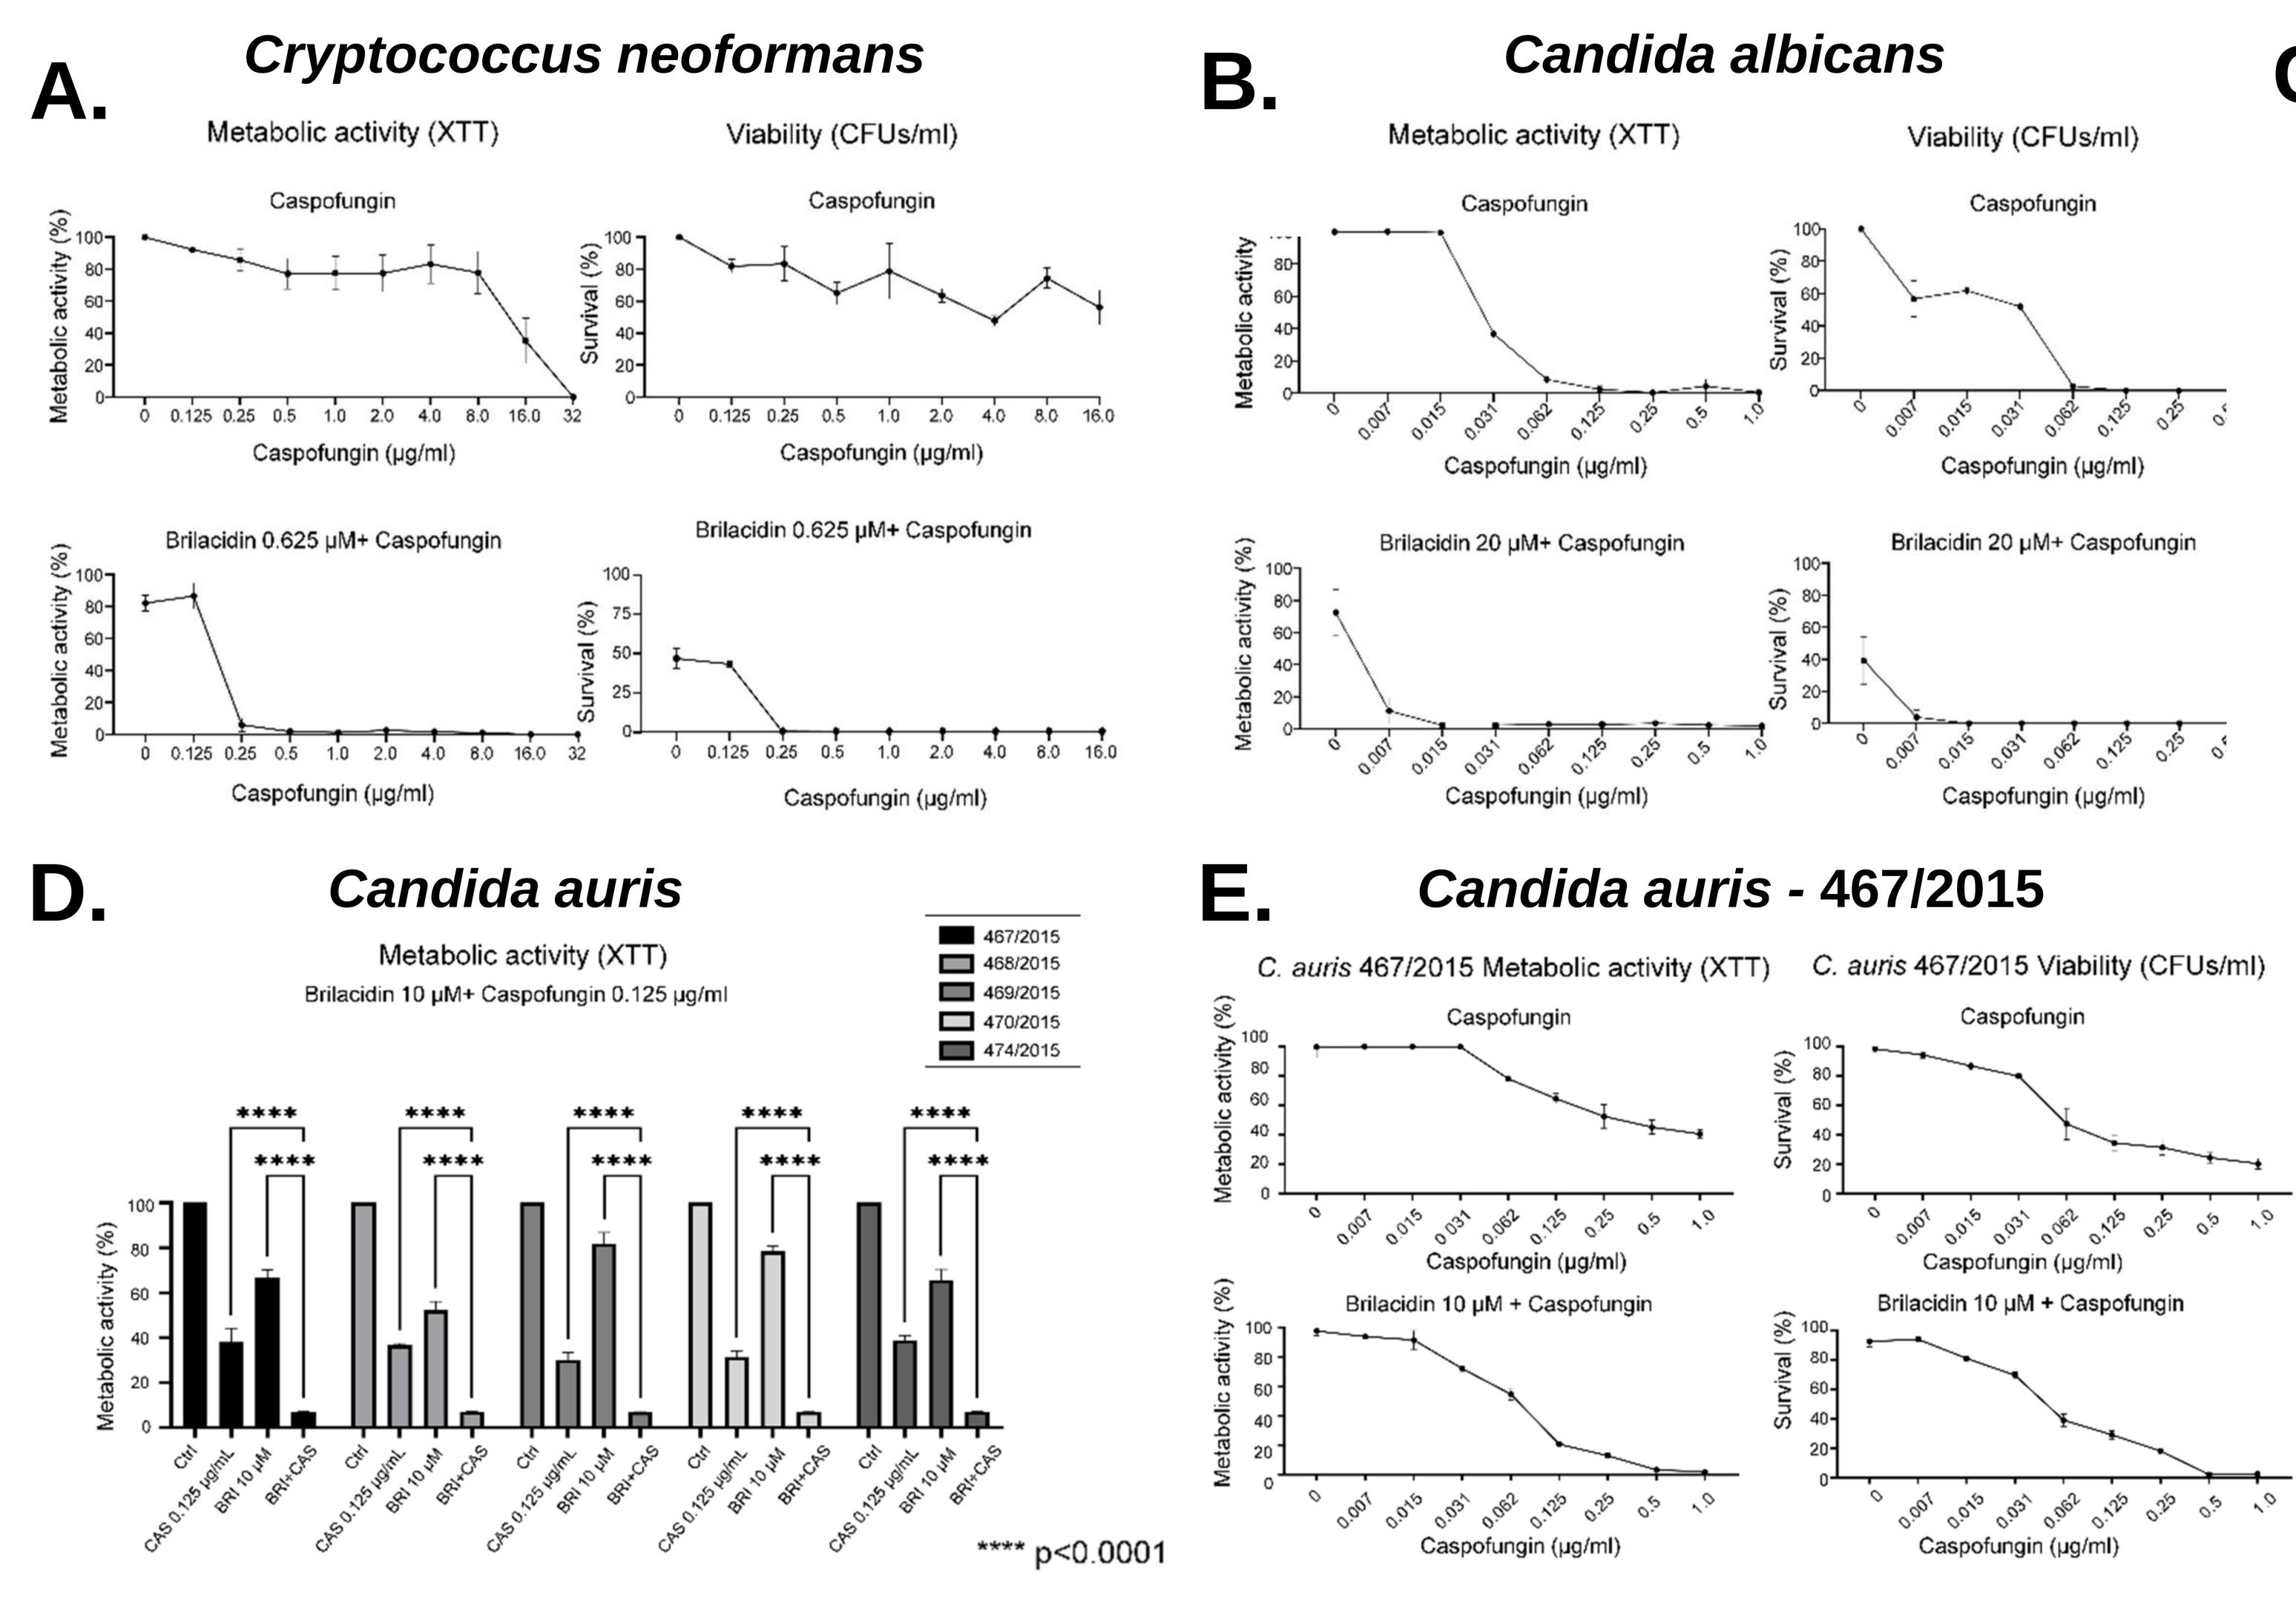

A.
Cryptococcus neoformans
Metabolic activity (XTT)
Viability (CFUs/ml)
Caspofungin
Brilacidin 0.625 µM+ Caspofungin
B.
Candida albicans
Metabolic activity (XTT)
Viability (CFUs/ml)
Brilacidin 20 µM+ Caspofungin
D.
Candida auris
Metabolic activity (XTT)
Brilacidin 10 µM+ Caspofungin 0.125 µg/ml
**** p<0.0001
E.
Candida auris - 467/2015
C. auris 467/2015 Metabolic activity (XTT)
C. auris 467/2015 Viability (CFUs/ml)
Brilacidin 10 µM + Caspofungin

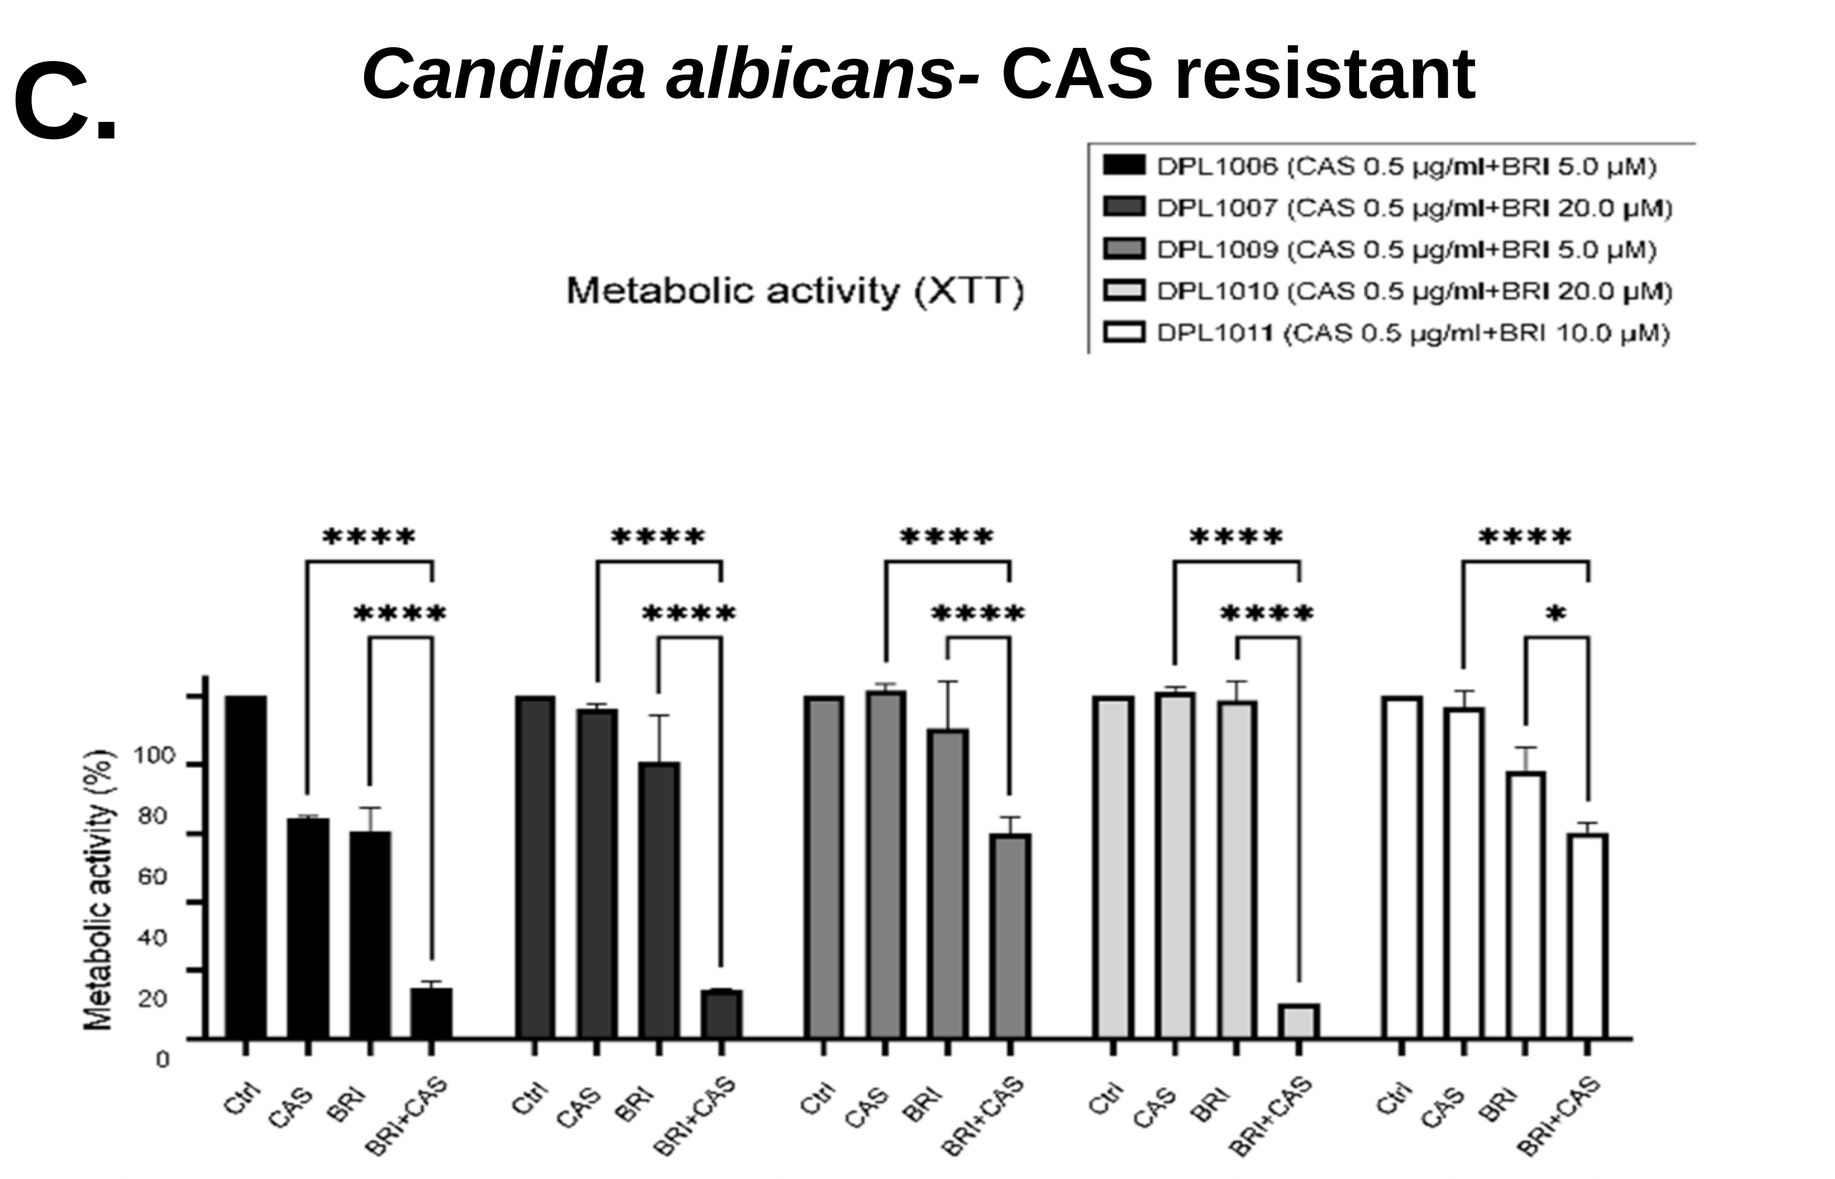

C.
Candida albicans- CAS resistant
Metabolic activity (XTT)
DPL1006 (CAS 0.5 µg/ml+BRI 5.0 µM)
DPL1007 (CAS 0.5 µg/ml+BRI 20.0 µM)
DPL1009 (CAS 0.5 µg/ml+BRI 5.0 µM)
DPL1010 (CAS 0.5 µg/ml+BRI 20.0 µM)
DPL1011 (CAS 0.5 µg/ml+BRI 10.0 µM)

Figure 4. BRI synergizes CAS against other human fungal pathogens. Metabolic activity expressed by XTT of **(A)** *C. neoformans,* **(B)** *C. albicans* and **(C)** *C. albicans* CAS-resistant strains grown for 48h in the absence or presence of CAS or BRI + CAS; **(D)** *C. auris* grown for 48h in the absence or presence of CAS, BRI or BRI + CAS; **(E)** *C. auris* 467/2015 strain grown for 48h in the absence or presence of CAS or BRI + CAS. Percentage of survival is expressed as CFU/mL of cells grown for 48h in the absence or presence of CAS or BRI + CAS. (*, $p < 0.05$, **, $p < 0.01$, ***, $p < 0.001$, and ****, $p < 0.0001$). All the results are the average of three repetitions ± standard deviation.

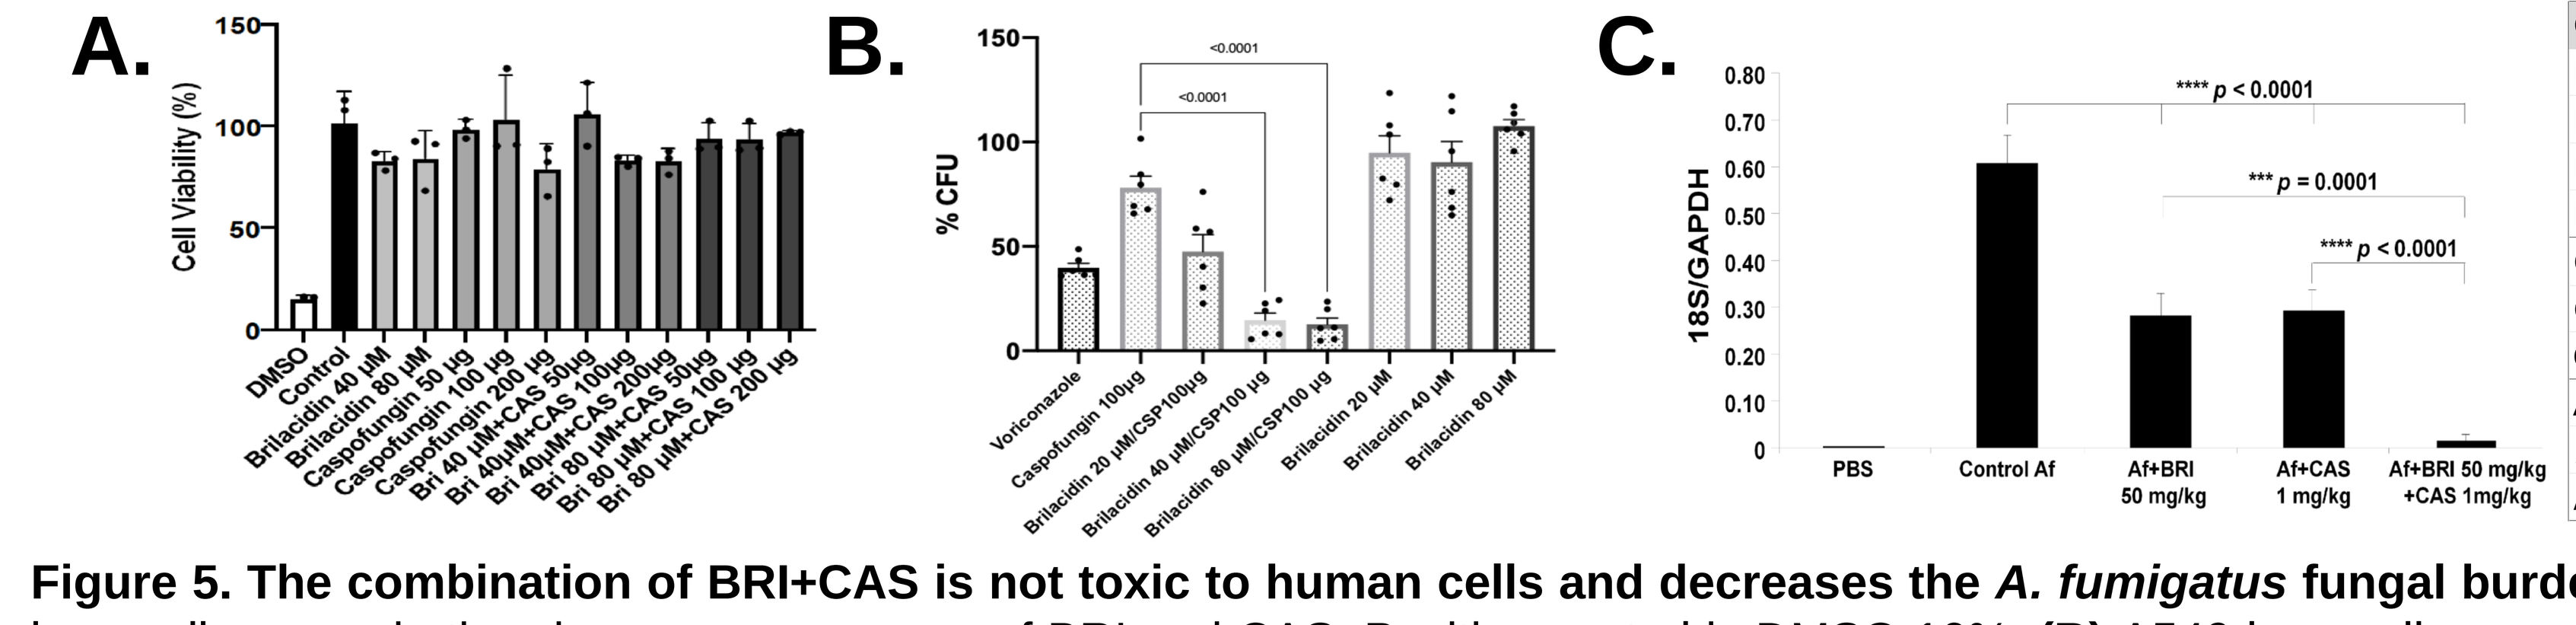

A.
Cell Viability (%)
B.
% CFU
C.
18S/GAPDH
**** p < 0.0001
*** p = 0.0001
**** p < 0.0001
PBS
Control Af
Af+BRI 50 mg/kg
Af+CAS 1 mg/kg
Af+BRI 50 mg/kg +CAS 1mg/kg

Comparisons test	Signific.	Summary	P Value
PBS *vs.* Control Af	Yes	****	<0.0001
PBS *vs.* Af + BRI	Yes	****	<0.0001
PBS *vs.* Af + CAS	Yes	****	<0.0001
PBS *vs.* Af + CAS + BRI	No	ns	0.9995
Control Af *vs.* Af + BRI	**Yes**	****	**<0.0001**
Control Af *vs.* Af + CAS	**Yes**	****	**<0.0001**
Control Af *vs.* Af + CAS + BRI	**Yes**	****	**<0.0001**
Af + BRI *vs.* Af + CAS	No	ns	0.9661
Af + BRI *vs.* Af + CAS + BRI	**Yes**	***	**0.0001**
Af + CAS *vs.* Af + CAS + BRI	**Yes**	****	**<0.0001**

Figure 5. The combination of BRI+CAS is not toxic to human cells and decreases the *A. fumigatus* fungal burden in a chemotherapeutic murine model. (A) A549 lung cells grown in the absence or presence of BRI and CAS. Positive control is DMSO 10%. **(B)** A549 lung cells were infected with *A. fumigatus* conidia in the presence or absence of BRI and CAS. In both (A) and (B), the percentage of cell viability is expressed as the absorbance value of each well/absorbance value of the control x 100. The results are the average of three repetitions ± standard deviation. **(C)** Fungal burden was determined 72h p.i. by real-time qPCR based on 18S rRNA gene of *A. fumigatus* and the mouse GAPDH gene. Fungal and mouse DNA quantities were obtained from the Ct values from an appropriate standard curve. The results are the means ± standard deviation of five animals. Statistical analysis was One-way ANOVA followed by Tukey's multiple comparison test.

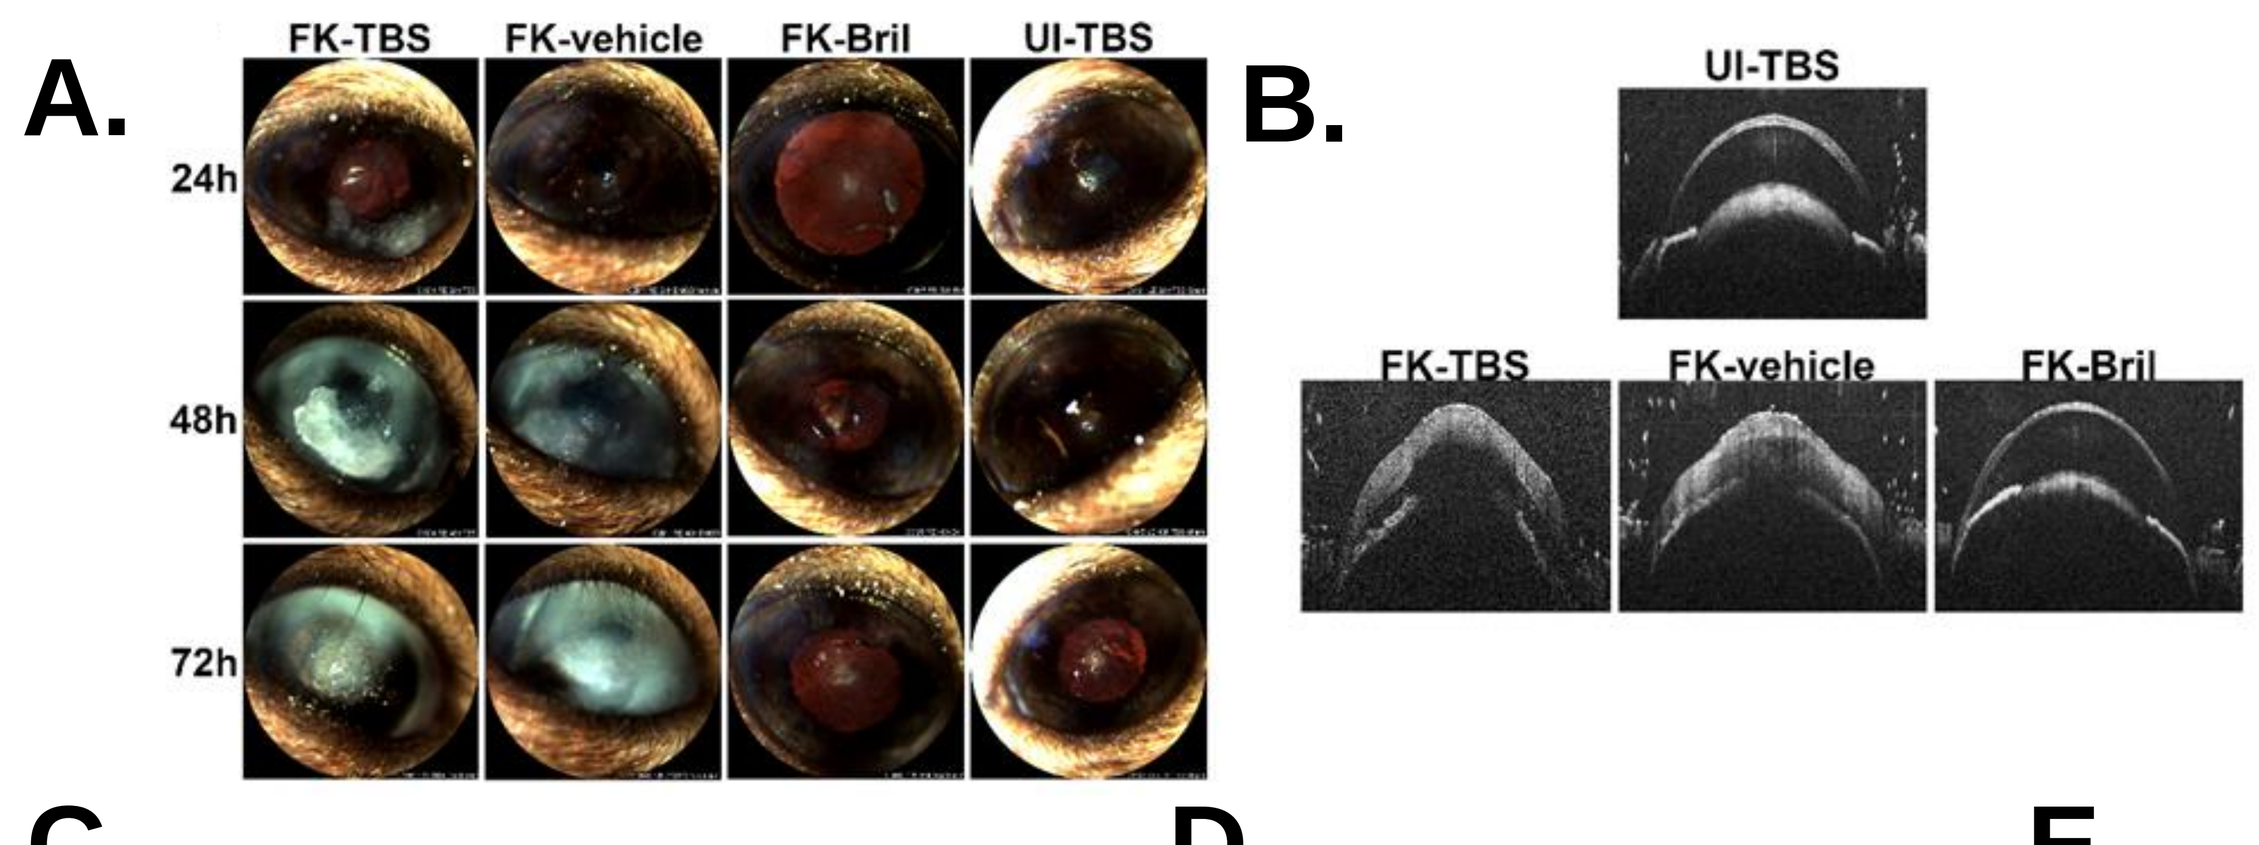

A.
FK-TBS
FK-vehicle
FK-Bril
UI-TBS
24h
48h
72h
B.
UI-TBS
FK-TBS
FK-vehicle
FK-Bril

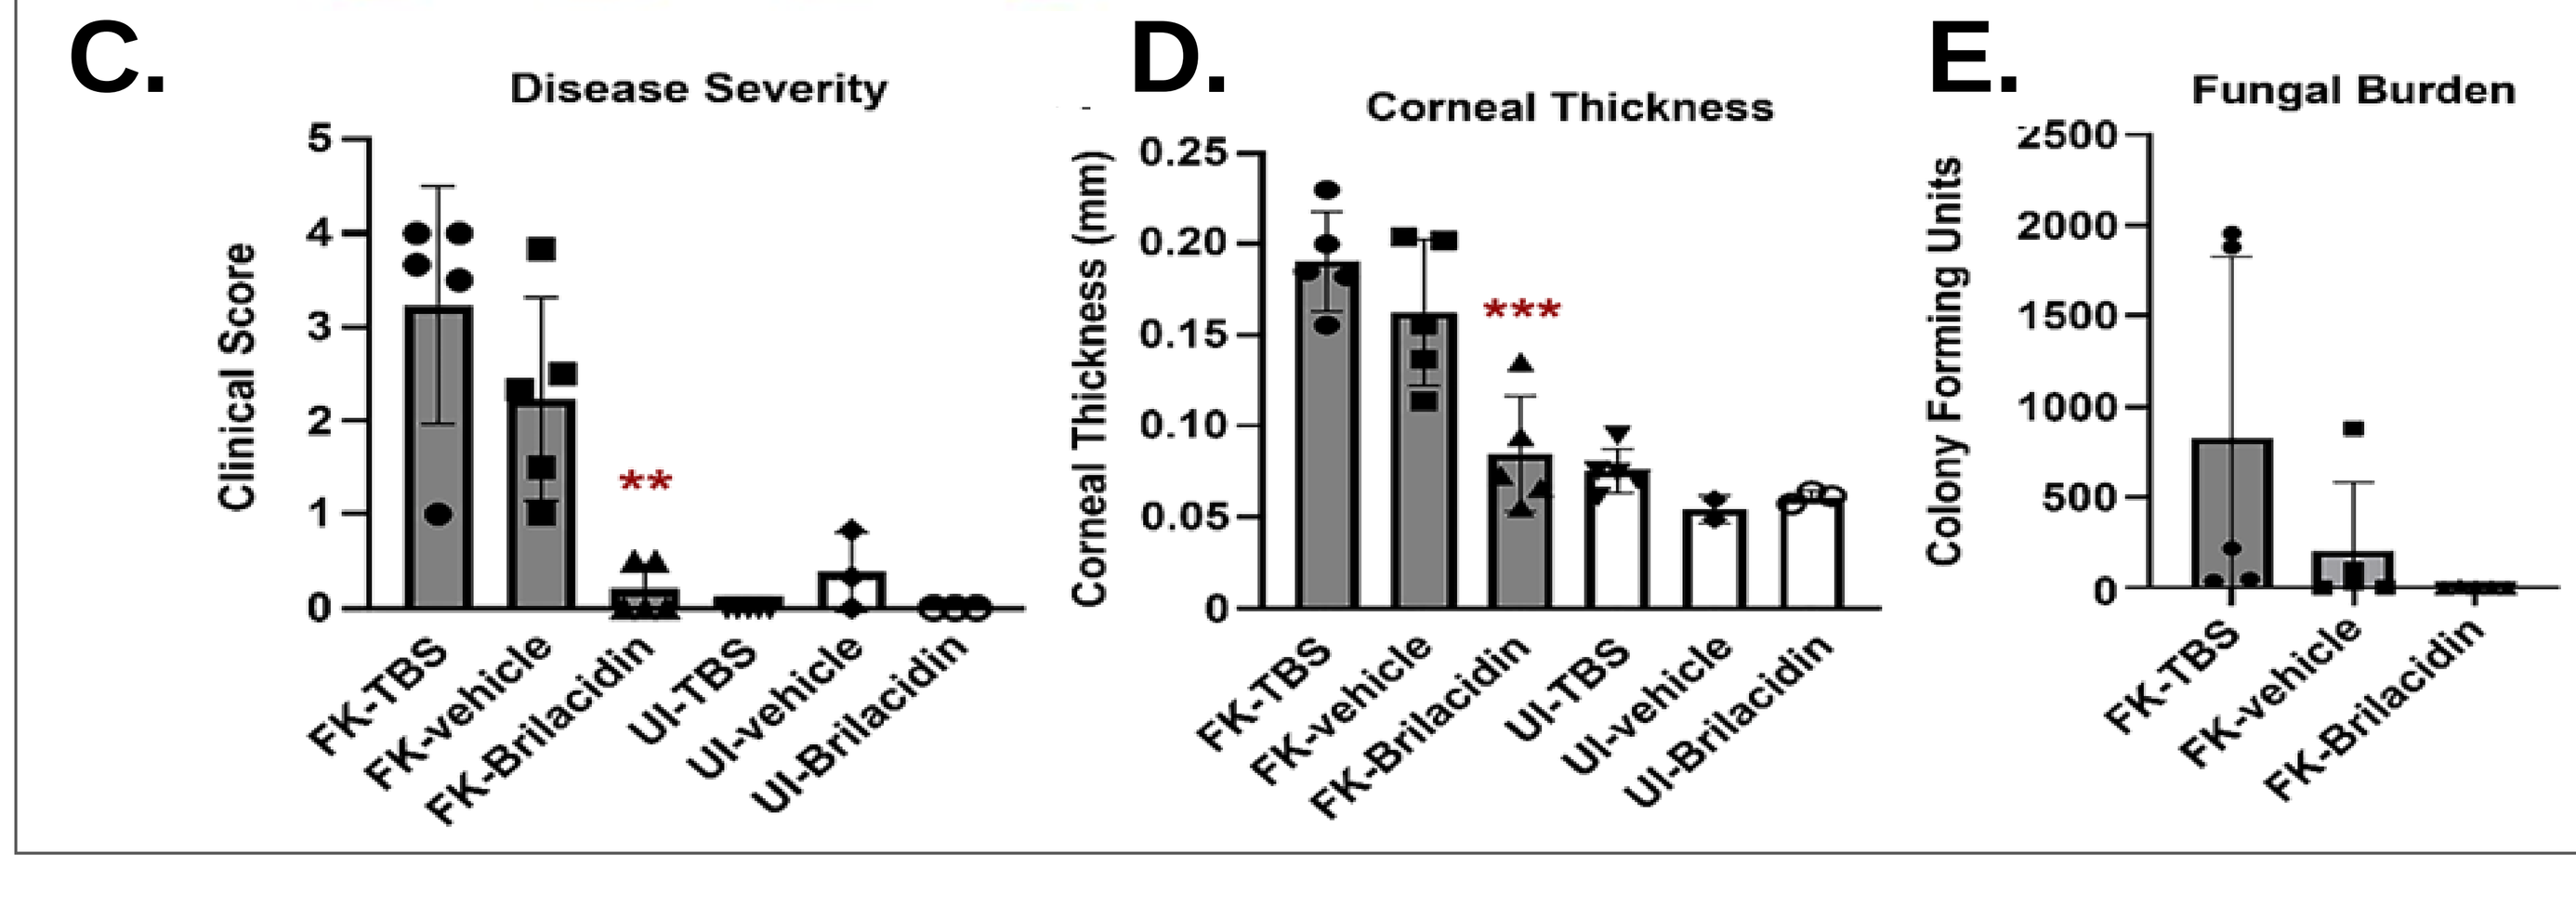

C.
Disease Severity
Clinical Score
FK-TBS
FK-vehicle
FK-Brilacidin
UI-TBS
UI-vehicle
UI-Brilacidin
D.
Corneal Thickness
Corneal Thickness (mm)
E.
Fungal Burden
Colony Forming Units

Figure 6. BRI blocks fungal growth and disease development in a murine model of fungal keratitis. (A) Representative images of slit-lamp images of *A. fumigatus*-infected murine corneas treated with TBS, DMSO (vehicle) or BRI (n=5/group). UI-TBS are uninfected (sham) corneas treated with TBS. **(B)** Representative ocular coherence tomography (OCT) scans of the indicated groups at 72h p.i. **(C)** Clinical scoring based on the slit-images in panel A. **(D)** Corneal thickness measurements based on the OCT analysis depicted in panel B. **(E)** CFU measurements from homogenized corneas taken 72h p.i. In each panel, the infected (FK) and UI groups were analyzed by one-way ANOVA; ** (p<.01), *** (p<.001).

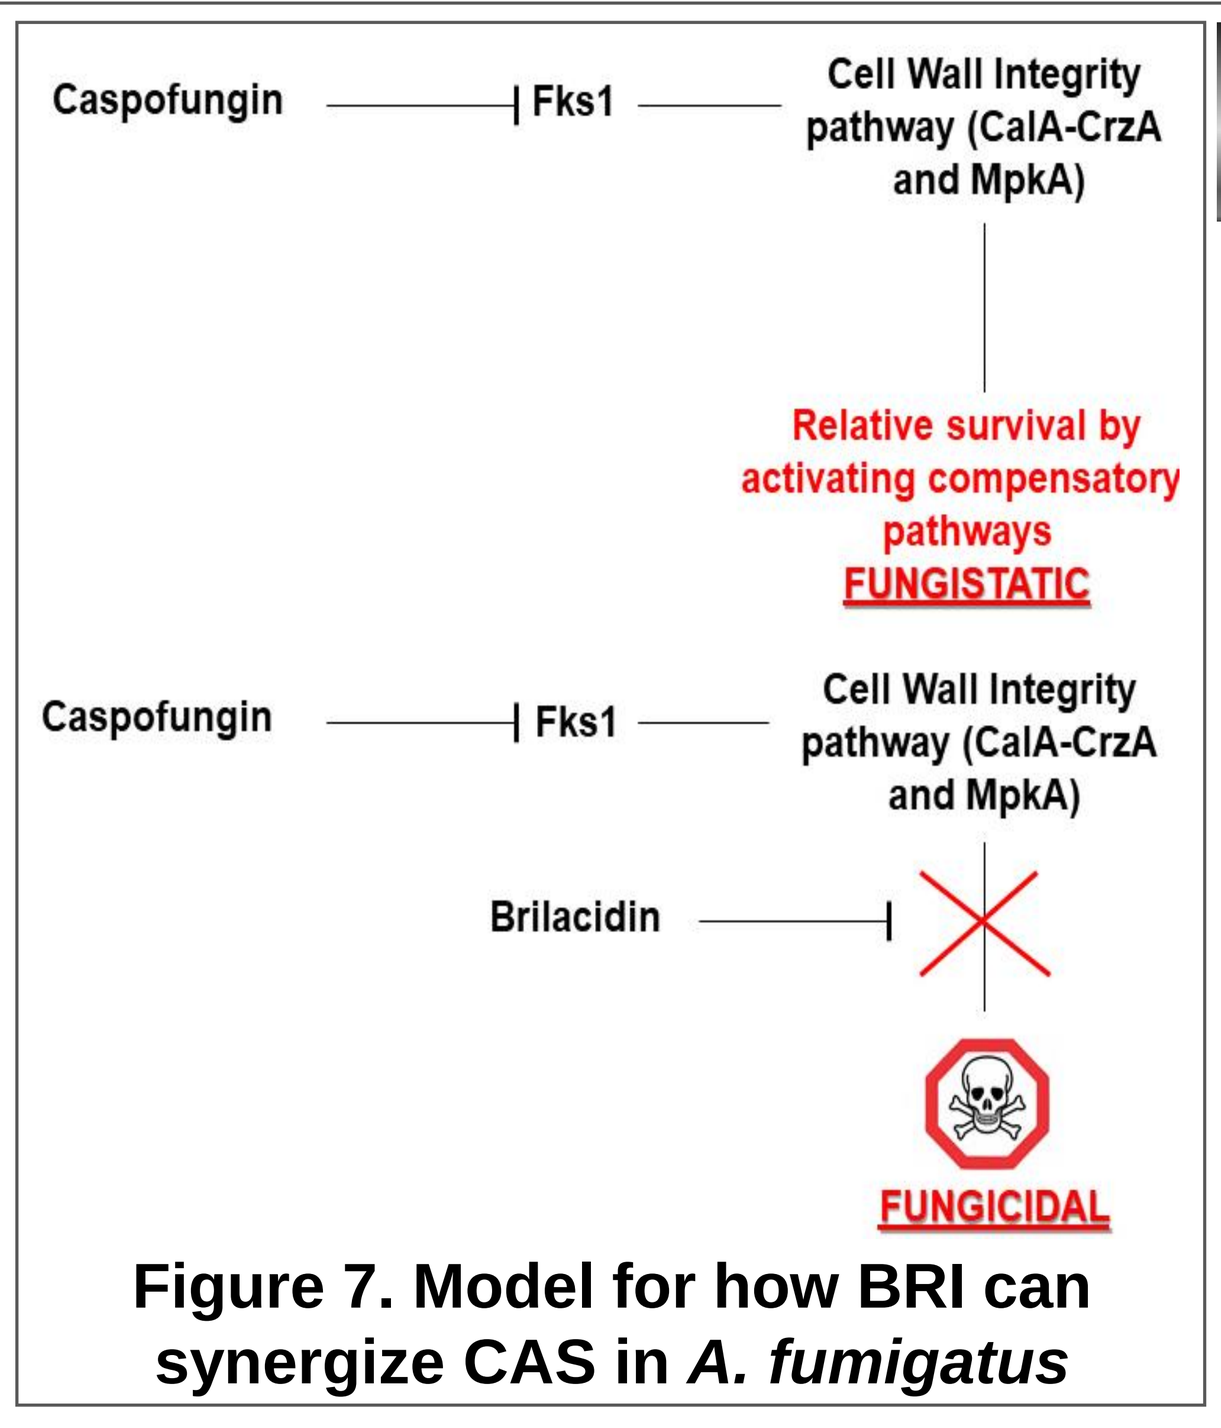

Caspofungin
Fks1
Cell Wall Integrity pathway (CalA-CrzA and MpkA)
Relative survival by activating compensatory pathways
FUNGISTATIC
Caspofungin
Fks1
Cell Wall Integrity pathway (CalA-CrzA and MpkA)
Brilacidin
FUNGICIDAL

Figure 7. Model for how BRI can synergize CAS in *A. fumigatus*

CONCLUSIONS

Here we identified the host defense peptide mimetic BRI as a potential synergizer of CAS. The combination CAS + BRI shows low cytotoxicity in an *in vitro* model of human cells and displayed high efficacy against several human fungal pathogens *in vitro*. In addition, the combination CAS + BRI was effective *in vivo* in (i) a murine model of fungal keratitis and (ii) a chemotherapeutic murine model of invasive aspergillosis. Taken together, BRI acts by impacting *A. fumigatus* calcineurin signaling and the cell wall integrity pathway converting the fungistatic action of CAS against *A. fumigatus* into a fungicidal drug.

Acknowledgements: We would like to thank the FAPESP (grants 2016/07870-9 and 2021/04977-5; G.H.G) and CNPq (grants 301058/2019-9 and 404735/2018-5; G.H.G.), both from Brazil, and the NIAID-NIH (grants R01AI153356, and R01AI063503) to ASI from the USA, for financial support for our research.