Brilacidin, a Host Defense Peptide Mimetic, is a Broad-Spectrum Countermeasure Strategy Against Acutely Infectious Viruses

Carol Anderson[1], Michael Barrera[1], Niloufar Boghdeh[1], Kyle Weston[2], Jane Harness[2], Aarthi Narayanan[1]

[1] George Mason University; [2] Innovation Pharmaceuticals

Introduction

Acutely infectious viruses, including those that are transmissible by the respiratory and aerosol routes, pose critical threats to the warfighter and the civilian population. Aerosol-transmissible pathogens, such as bunyaviruses (Rift Valley Fever Virus [RVFV]), and alphaviruses (Venezuelan Equine Encephalitis Virus [VEEV], Eastern Equine Encephalitis Virus [EEEV]), have a broad range of host tropism, retain high rates of infectivity as aerosols, attain high viral load in the host over short periods of time, cause damage to the blood brain barrier (BBB), impact neurological integrity and are likely to contribute to organ-damage due to extreme inflammation. These viruses pose nontrivial challenges to the warfighter because no FDA-approved therapeutics or vaccines are currently available that can be rapidly scaled up, field-deployed and potentially mitigate the deleterious consequences of inflammation in addition to decreasing viral load. The long-term neurological sequelae that ensue from these acute infections, especially by the aerosol route, can lead to a life-long health burden for the warfighter. These pathogens are also naturally transmitted by mosquito vectors and are known to cause zoonotic disease in animals in addition to affecting humans on an annual basis in the United States and in other parts of the world. There is an urgent, unmet need for broad-spectrum intervention strategies that can ideally control the disease manifestations in the host and the spread of disease as a prophylactic countermeasure. Our ongoing studies with brilacidin, a host defense peptide (HDP)-based mimetic, have successfully demonstrated that brilacidin is able to interfere with viral integrity and exert an antiviral effect in vitro against candidate alphaviruses and bunyavirus. Furthermore, early indications support the potential of brilacidin to also act in an anti-inflammatory capacity by its impact on inflammatory cytokine expression.

Methods

Appropriate cell lines (Vero cells, U87MG cells, HSAE cells and HepG2 cells) were infected with RVFV (MP-12 strain), VEEV (TC-83 and TrD strains) and SINV +/- brilacidin. Culture supernatants and nucleic acid lysates were quantified for extracellular and intracellular viral load and impact on infectivity by plaque and qRT-PCR assays. Impact of brilacidin on cell viability and lack of toxicity were ascertained by CellTiter Glo assay. Impact of treatment on inflammatory events were quantified by a combination of PCR (gene expression) and ELISA (protein expression). Early studies involving cell biological mechanisms have involved transmission electron microscopy of virus distribution in infected cells +/- brilacidin.

Results

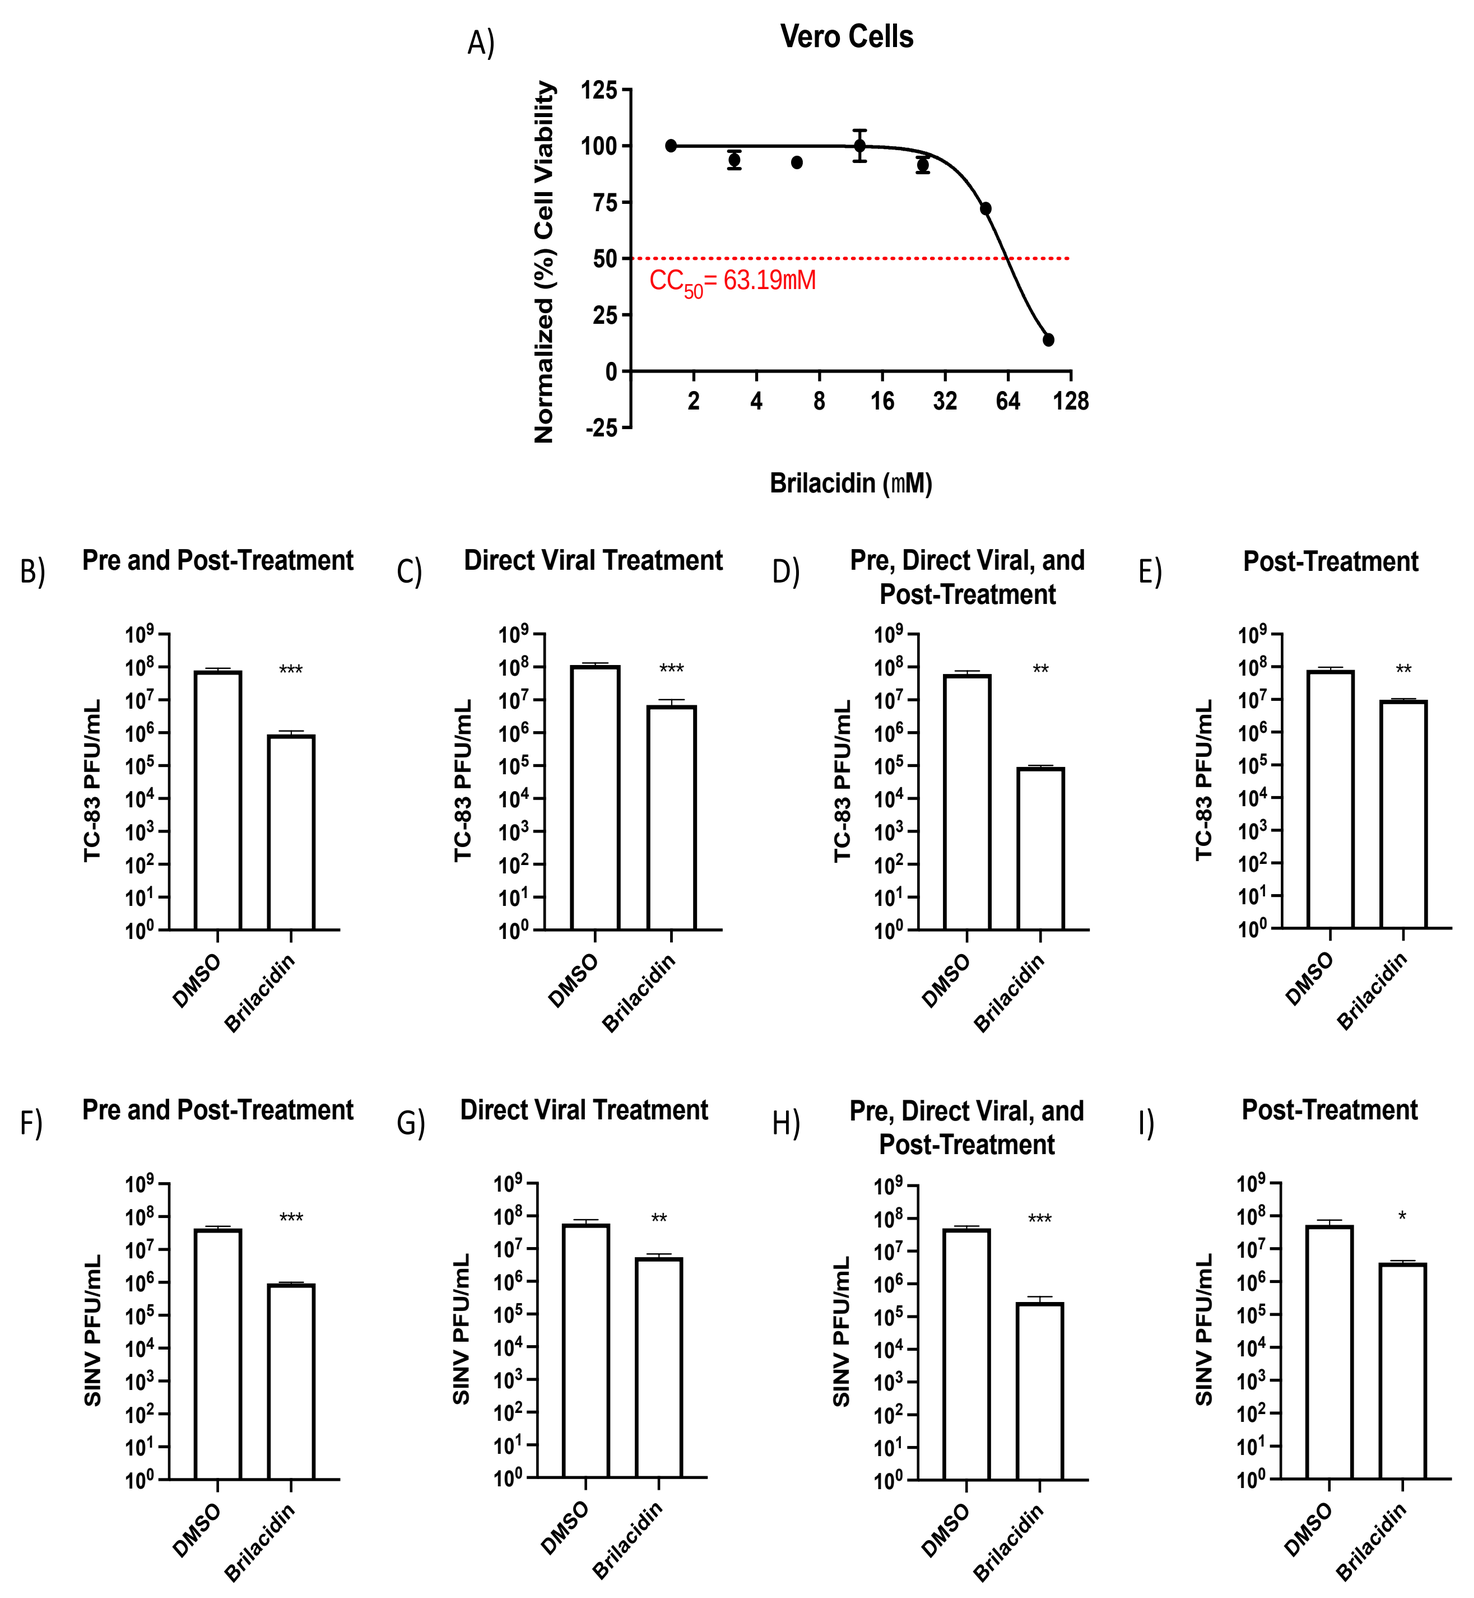
A) Vero Cells
Normalized (%) Cell Viability
CC_{50}= 63.19mM
Brilacidin (mM)
B) Pre and Post-Treatment
C) Direct Viral Treatment
D) Pre, Direct Viral, and Post-Treatment
E) Post-Treatment
TC-83 PFU/mL
F) Pre and Post-Treatment
G) Direct Viral Treatment
H) Pre, Direct Viral, and Post-Treatment
I) Post-Treatment
SINV PFU/mL
DMSO
Brilacidin

Figure 1. Brilacidin inhibits alphavirus replication in Vero cells. (A) Cytotoxicity of brilacidin was determined in Vero cells after 24hrs of treatment. (B-I) Effects of brilacidin treatments on VEEV TC-83 and SINV. (B,F) Cells were pretreated with brilacidin (20µM) for one hour and then infected with VEEV TC-83 or SINV (MOI of 0.1) for one hour. After infection, brilacidin (20µM) containing media was applied. (C,G) VEEV TC-83 and SINV were directly treated with brilacidin at 20µM for 1 hour prior to infection. After infection, inoculum was removed, and media was applied. (D,H) Treatments described in (B,F) and (C,G) were combined. (E,I) Cells were infected with VEEV TC-83 or SINV (MOI of 0.1) for one hour. After infection, the inoculum was removed and brilacidin (20µM) containing media was applied. All supernatants were collected at 18hpi, and the viral titer was determined by plaque assay. * $p < 0.05$, ** $p < 0.01$, *** $p < 0.001$, **** $p < 0.0001$.

Figure 2: Bunyavirus data

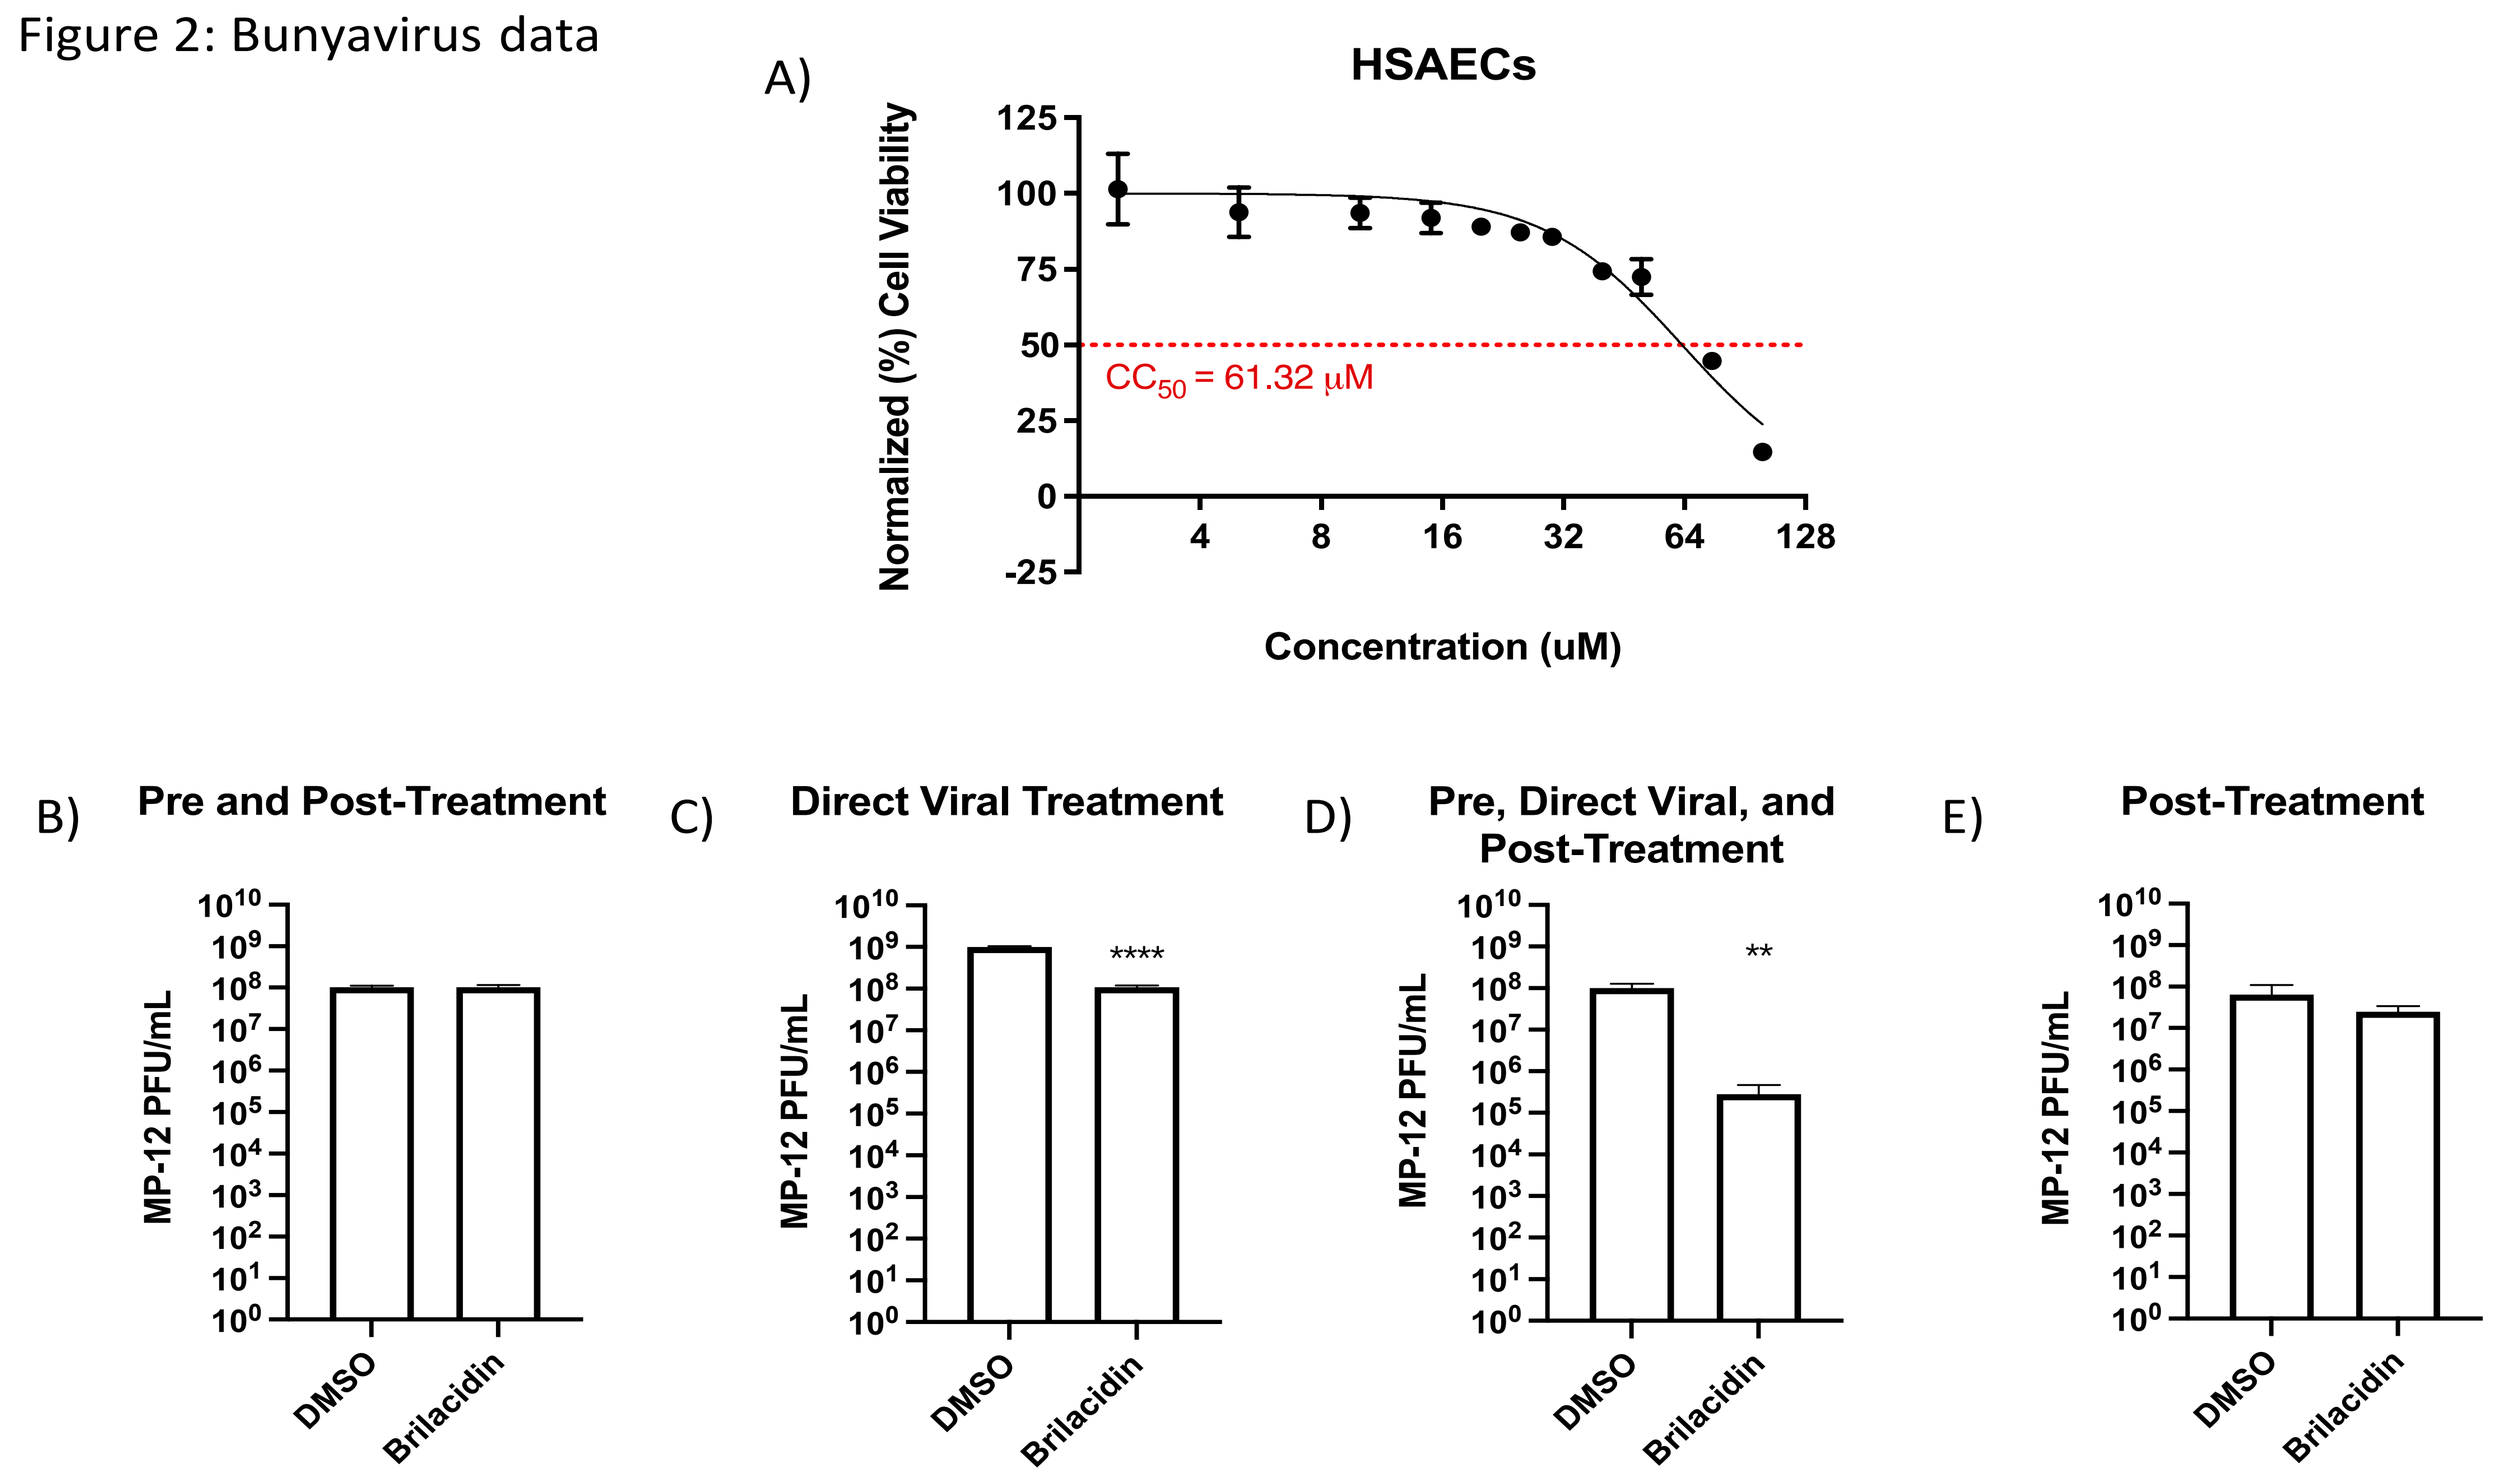
A) HSAECs
Normalized (%) Cell Viability
CC_{50} = 61.32 µM
Concentration (uM)
B) Pre and Post-Treatment
C) Direct Viral Treatment
D) Pre, Direct Viral, and Post-Treatment
E) Post-Treatment
MP-12 PFU/mL
DMSO
Brilacidin

Figure 2. Brilacidin inhibits Rift Valley Fever Virus (RVFV MP-12) replication in Human Small Airway Epithelial cells (HSAECs). (A) Cytotoxicity of brilacidin was determined in HSAEC cells using CellTiter-Glo Luminescent Cell Viability Assay after 24hrs of treatment. (B-E) Effects of various brilacidin treatments on RVFV MP-12. (B) Cells were pretreated with brilacidin (20µM) for one hour and then infected with MP-12 (MOI of 0.1) for one hour. After infection, brilacidin (20µM) containing media was applied. (C) MP-12 was directly treated with brilacidin at 20µM for 1 hour prior to infection. After infection, inoculum was removed, and media was applied. (D) Treatments described in (B) and (C) were combined. (E) Cells were infected with MP-12 (MOI of 0.1) for one hour. After infection, the inoculum was removed and brilacidin (20µM) containing media was applied. All supernatants were collected at 16hpi, and the viral titer was determined by plaque assay. Values are an average of 3 biological replicates ± standard deviation. ** $p < 0.01$, **** $p < 0.0001$.

Figure 3: Alphavirus and bunyavirus – virulent strains

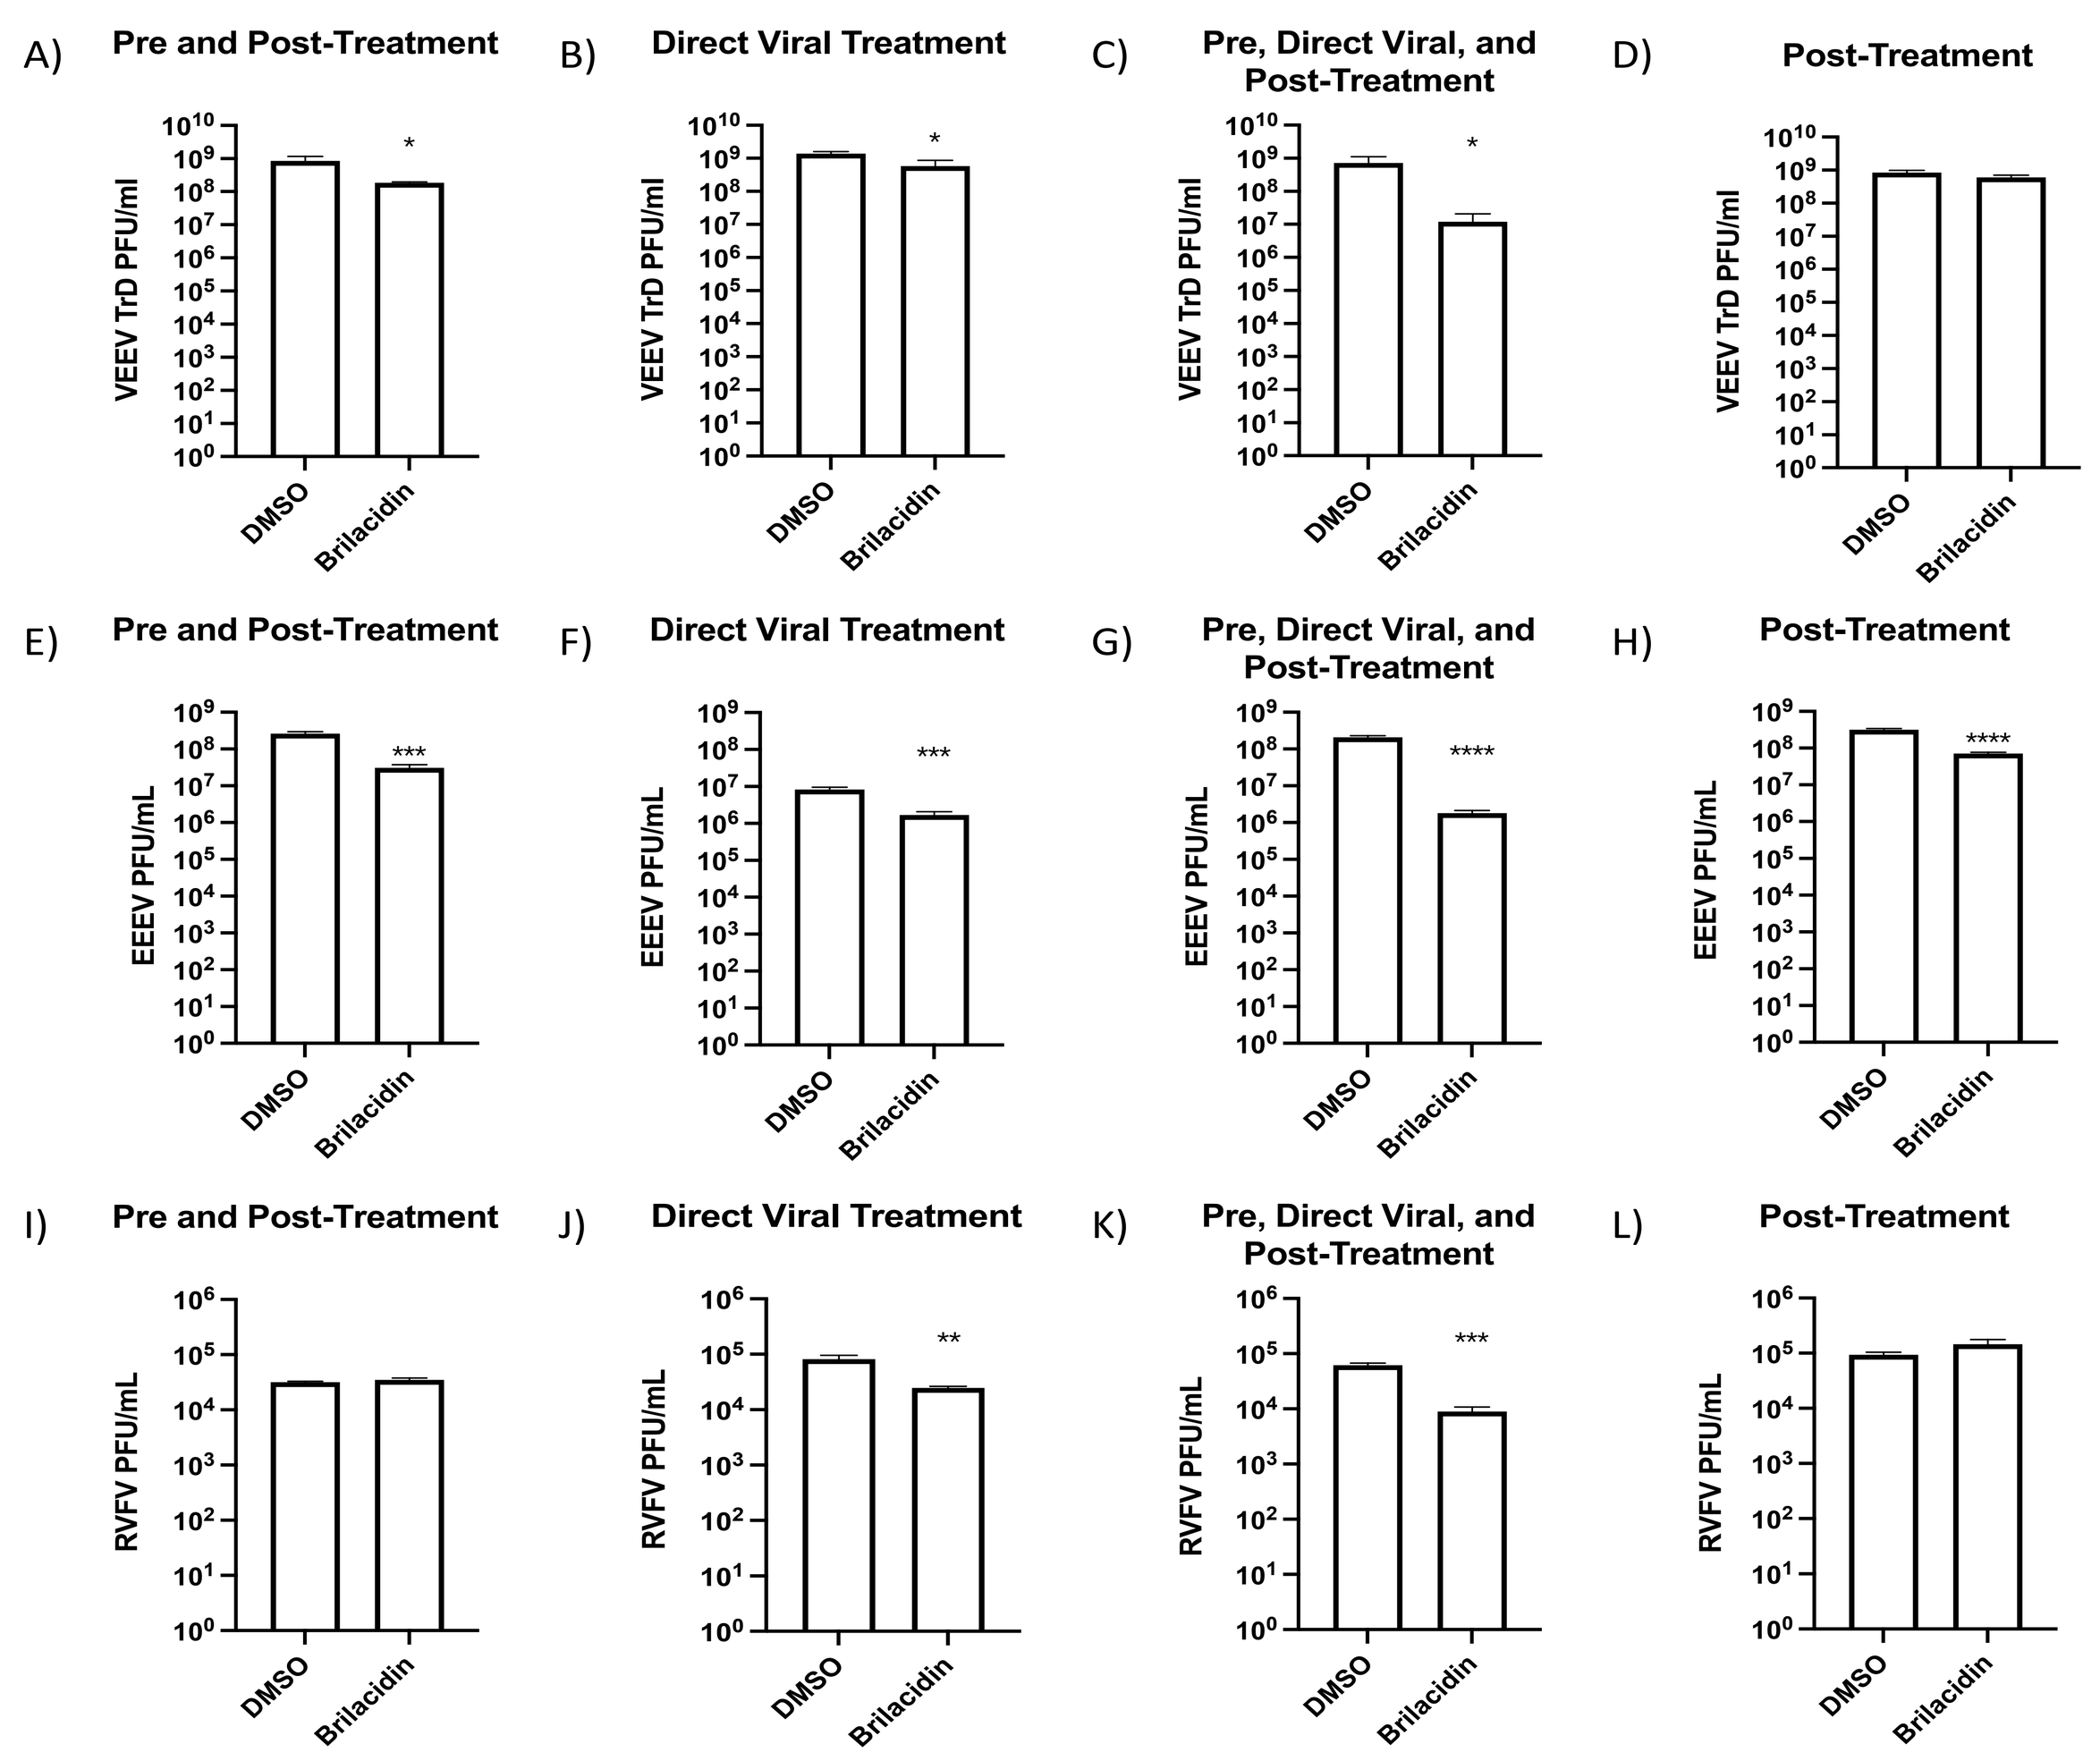
A) Pre and Post-Treatment
B) Direct Viral Treatment
C) Pre, Direct Viral, and Post-Treatment
D) Post-Treatment
VEEV TrD PFU/ml
E) Pre and Post-Treatment
F) Direct Viral Treatment
G) Pre, Direct Viral, and Post-Treatment
H) Post-Treatment
EEEV PFU/mL
I) Pre and Post-Treatment
J) Direct Viral Treatment
K) Pre, Direct Viral, and Post-Treatment
L) Post-Treatment
RVFV PFU/mL
DMSO
Brilacidin

Figure 3. Brilacidin inhibits fully virulent alphaviruses and bunyavirus. (A-L) Effects of various brilacidin treatments on VEEV TrD, EEEV FL93-939, and RVFV ZH501. (A,E,I) Cells were pretreated with brilacidin (20µM) for one hour and then infected (MOI of 0.1) for one hour. After infection, brilacidin (20µM) containing media was applied. (B,F,J) Viruses were directly treated with brilacidin at 20µM for 1 hour prior to infection. After infection, inoculum was removed, and media was applied. (C,G,K) Treatments described in (A) and (B) were combined. (D,H,L) Cells were infected (MOI of 0.1) for one hour. After infection, the inoculum was removed and brilacidin (20µM) containing media was applied. VEEV and EEEV supernatants were collected at 18hpi and RVFV was collected at 16hpi. Viral titer was determined by plaque assay. Values are an average of 3 biological replicates ± standard deviation. * $p < 0.05$, ** $p < 0.01$, *** $p < 0.001$, **** $p < 0.0001$.

Figure 4: Echovirus

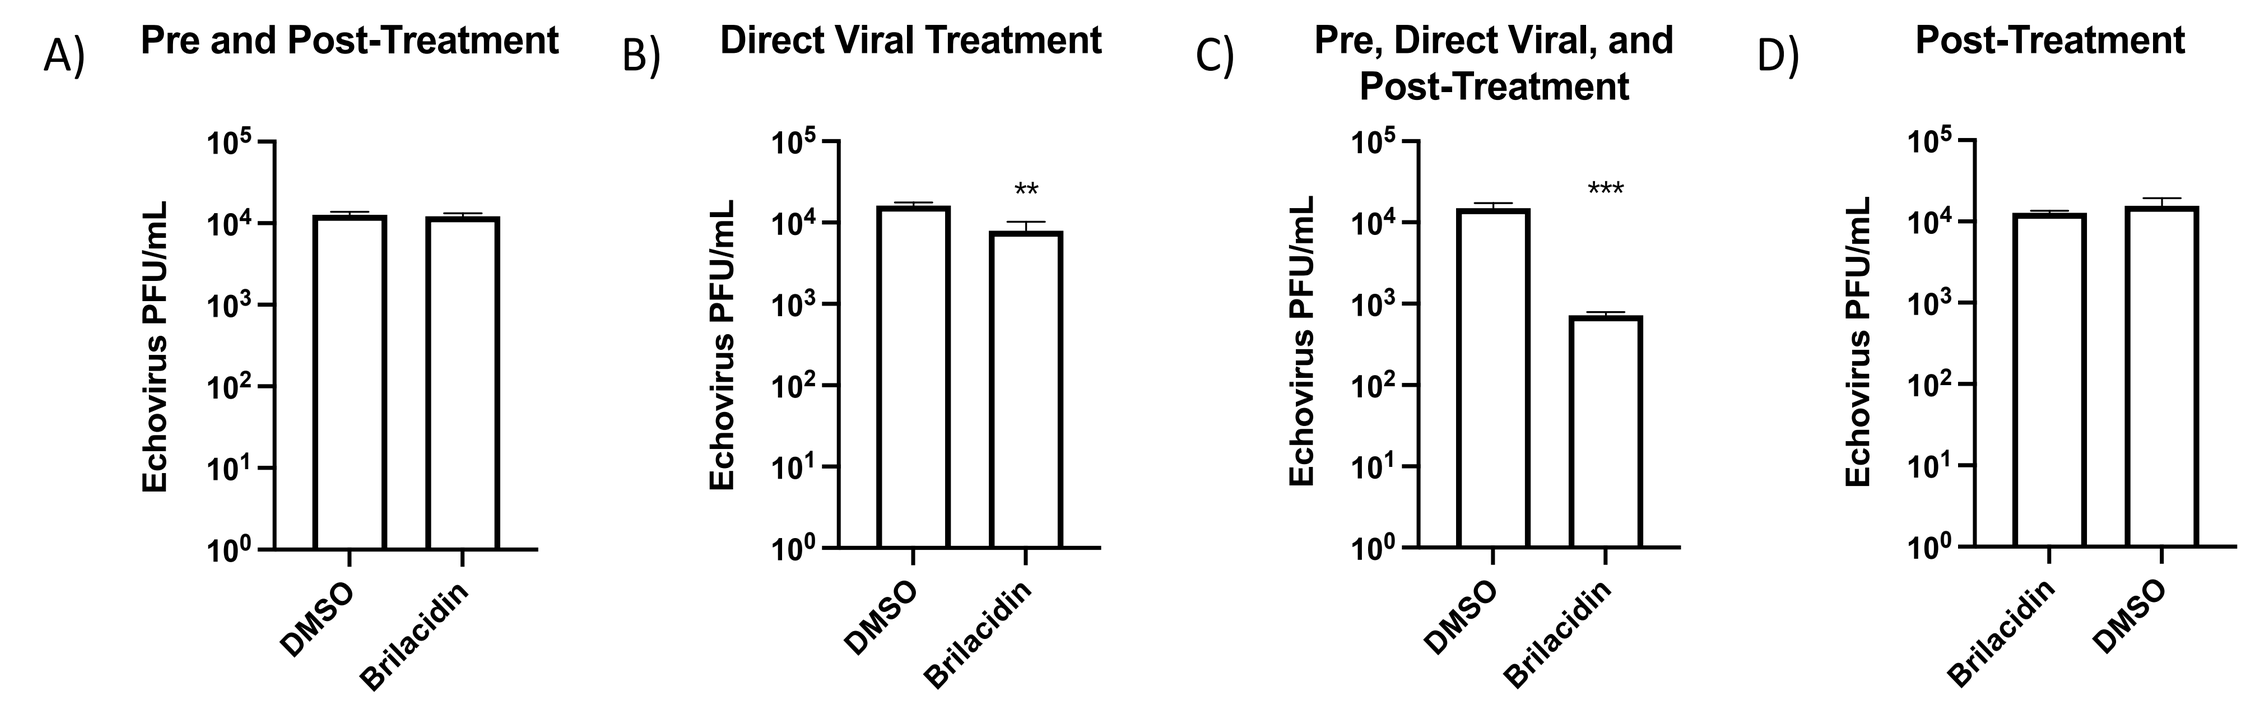
A) Pre and Post-Treatment
B) Direct Viral Treatment
C) Pre, Direct Viral, and Post-Treatment
D) Post-Treatment
Echovirus PFU/mL
DMSO
Brilacidin

Figure 4. Brilacidin inhibits Echovirus replication Vero cells. (A-D) Effects of various brilacidin treatments on Echovirus. (A) Cells were pretreated with brilacidin (20µM) for one hour and then infected (MOI of 0.1) for one hour. After infection, brilacidin (20µM) containing media was applied. (B) Echovirus was directly treated with brilacidin at 20µM for 1 hour prior to infection. After infection, inoculum was removed, and media was applied. (C) Treatments described in (A) and (B) were combined. (D) Cells were infected (MOI of 0.1) for one hour. After infection, the inoculum was removed and brilacidin (20µM) containing media was applied. All supernatants were collected at 24hpi, and the viral titer was determined by plaque assay. Values are an average of 3 biological replicates ± standard deviation. ** $p < 0.01$, *** $p < 0.001$.

Discussion/Conclusion

The observations that we continue to make in experiments involving brilacidin in the context of acutely infectious viruses support the idea of brilacidin functioning in a broad-spectrum capacity as an antiviral compound. Several lines of evidence suggest that brilacidin may affect virion integrity and hence impact viral entry, thus positioning it well as a broad-spectrum prophylactic countermeasure solution. The observations about anti-inflammatory activities (data not shown) indicate that intracellular events are also modulated by brilacidin treatment. Brilacidin is thus able to exert a combined protective effect by decreasing both viral and inflammatory load. Brilacidin has undergone several clinical studies as treatment for bacterial infections. Ongoing efforts are focused on dosing strategies for brilacidin to improve in vivo effectiveness in the form of a nasally-delivered countermeasure.

Acknowledgments

We would like to thank Innovation Pharmaceuticals for providing brilacidin to support all aspects of this study.